ribometrix

eIF4E inhibition exhibits anti-tumor activity and re-sensitizes acquired resistant KRAS^{G12C} NSCLC to KRAS inhibitors

Andrew S. Truong, Matthew B. Friedersdorf, Sarah E.G. Thompson, Roheeth K. Pavana, Ramamurty V.S. Changalvala, Jessica A. Sorrentino
Ribometrix Inc., Durham, NC

#202P

Abstract

The approval of KRAS^{G12C} selective inhibitors, sotorasib and adagrasib, marked a critical point in the effort to treat KRAS^{G12C} mutant NSCLC. Despite the initial clinical benefit observed after treatment, acquired resistance is observed in most patients representing an unmet medical need.

Eukaryotic translation initiation factor 4E (eIF4E) is a critical regulatory node for multiple oncogenic signaling pathways downstream of KRAS, including MAPK and PI3K pathways, making it a promising target to overcome resistance to KRAS^{mut} targeted therapies. Many bypass resistance mechanisms to KRAS inhibitors reactivate eIF4E, to selectively promote translation of pro-oncogenic and anti-apoptotic factors including Cyclin D1/3, BCL2, and MCL1.

Here, we present the development of a potent and selective eIF4E inhibitor, RBX-6610, for use in both KRAS^{G12C} targeted treatment-naïve and resistance settings. RBX-6610 elicits a reversible, dose-dependent cell cycle arrest. However, unlike targeting eIF4A, eIF4E inhibition selectively regulates translation of cancer-dependent pathway proteins instead of global protein synthesis, increasing tolerability. Additionally, RBX-6610 demonstrates consistent anti-proliferative effects in both sotorasib/adagrasib-naïve and resistant cell lines.

Combining RBX-6610 and KRAS inhibitors *in vitro* results in the marked increase of caspase 3/7 activity and cleaved PARP. Furthermore, the combination treatment not only produces synergistic apoptotic responses in naïve tumor cells but also re-sensitizes resistant cells to KRAS inhibitors. Importantly, RBX-6610 monotherapy *in vivo* causes significant tumor growth inhibition, and the combination therapy with sotorasib results in significant tumor regression in a naïve NSCLC xenograft model. A similar *in vivo* study, performed in a model of sotorasib acquired resistance validates RBX-6610's ability to re-sensitize tumor cells to KRAS inhibition.

Collectively, these data support the addition of RBX-6610 to standard of care in both the naïve and treatment resistant settings in KRAS^{mut} NSCLC patients. IND-enabling studies are planned, marking a significant step toward advancing RBX-6610, a potent eIF4E inhibitor, into the clinic.

Introduction

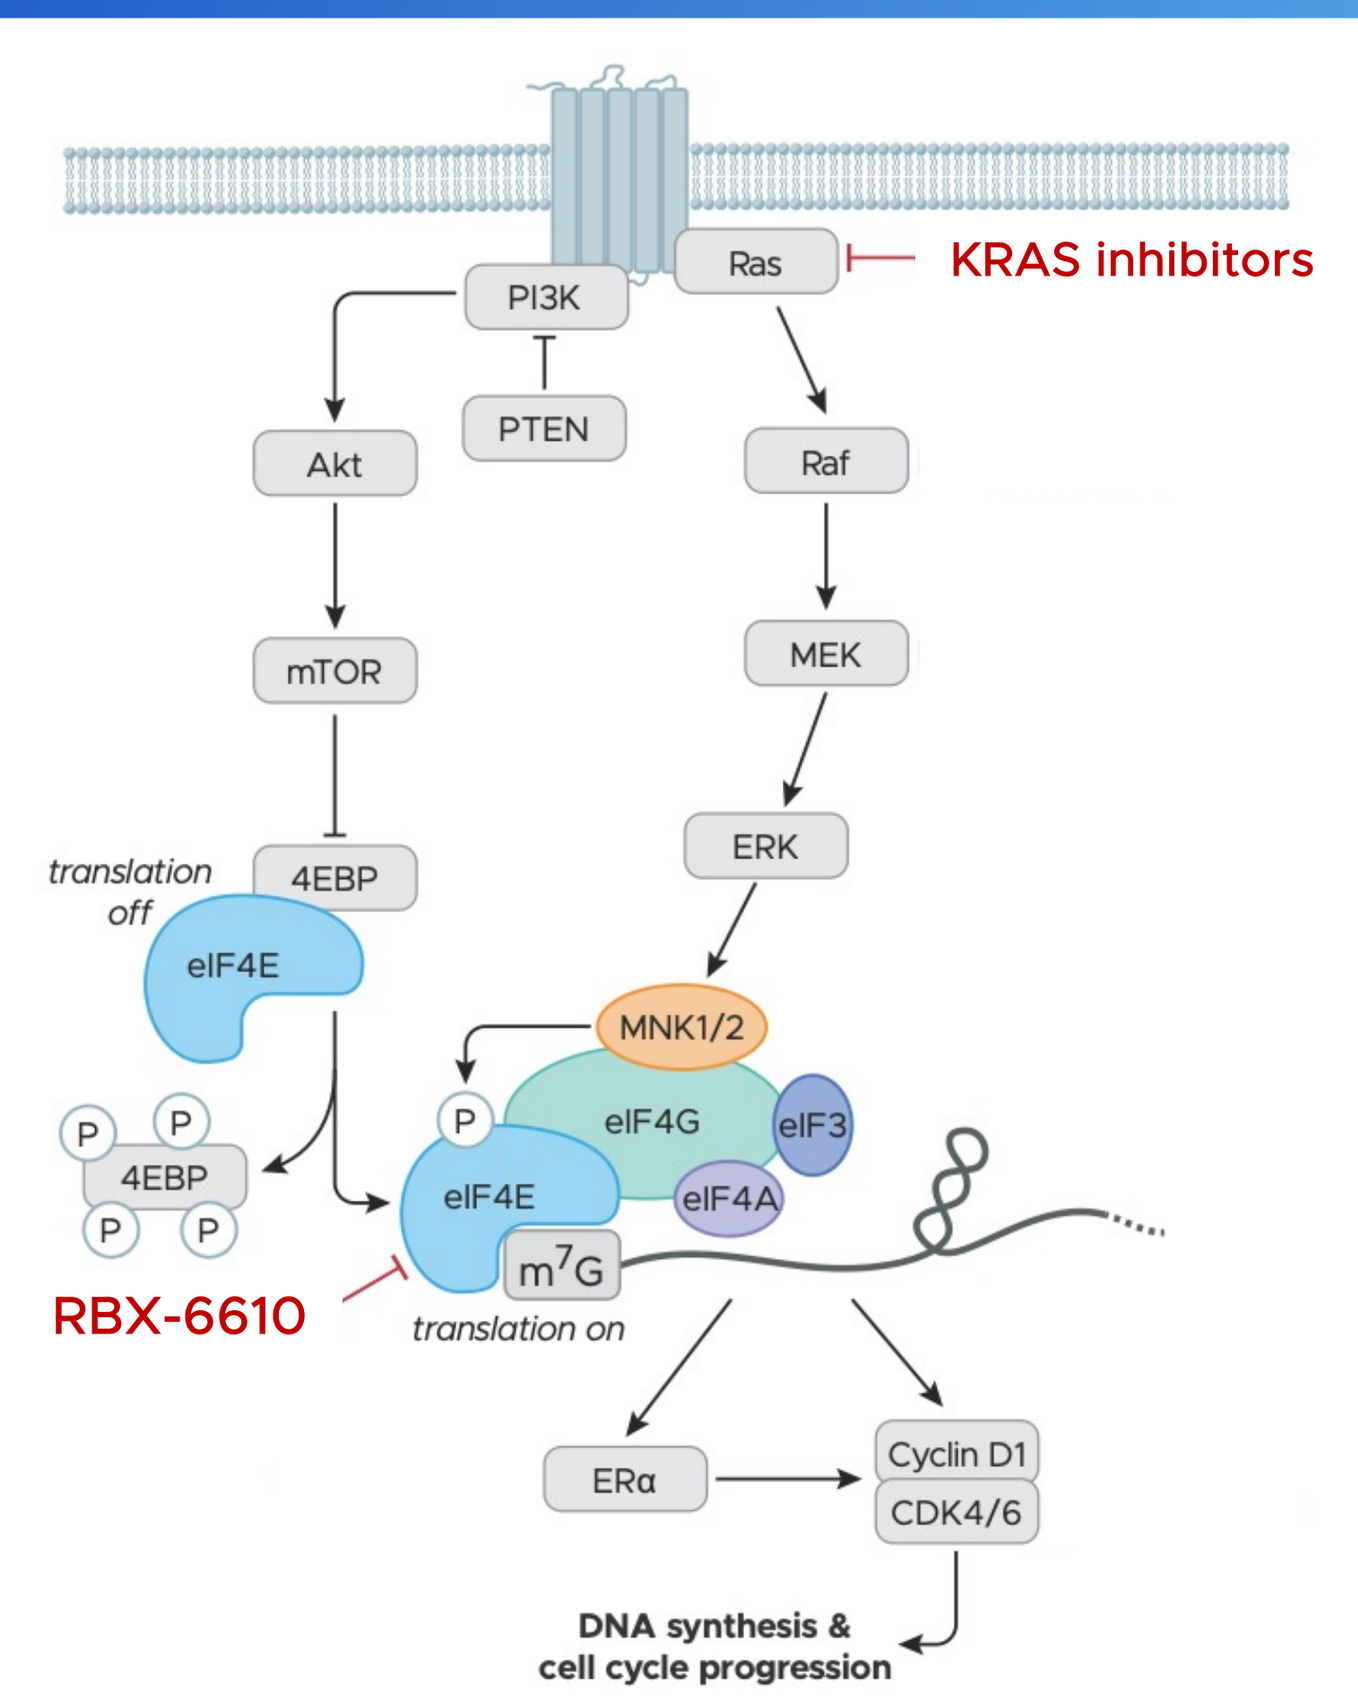
KRAS inhibitors
PI3K
Ras
PTEN
Akt
Raf
mTOR
MEK
ERK
translation off
4EBP
eIF4E
MNK1/2
eIF4G
eIF3
eIF4A
m7G
RBX-6610
translation on
ERα
Cyclin D1
CDK4/6
DNA synthesis & cell cycle progression

- eIF4E binds the 5′ cap of mRNA and promotes translation initiation. It is the rate-limiting factor for regulating protein synthesis
- eIF4E selectively regulates the translation of oncogenic mRNAs with highly structured 5'UTRs such as Cyclin D1, ODC1, and Myc
- eIF4E expression is elevated in many cancers, including lung cancer, and high levels are associated with poor prognosis
- Forced overexpression of eIF4E drives tumors in animal models, while genetic inhibition of eIF4E decreases tumor viability but not viability of normal cells
- eIF4E is a point of convergence for multiple pro-oncogenic signaling pathways, including Ras/Raf/MEK and PI3K/AKT/mTOR pathways
- Many resistance mechanisms for Ras/Raf/MEK pathway inhibitors result in hyper-activation of eIF4E

RBX-6610, an eIF4E inhibitor, blocks translation in NSCLC cells

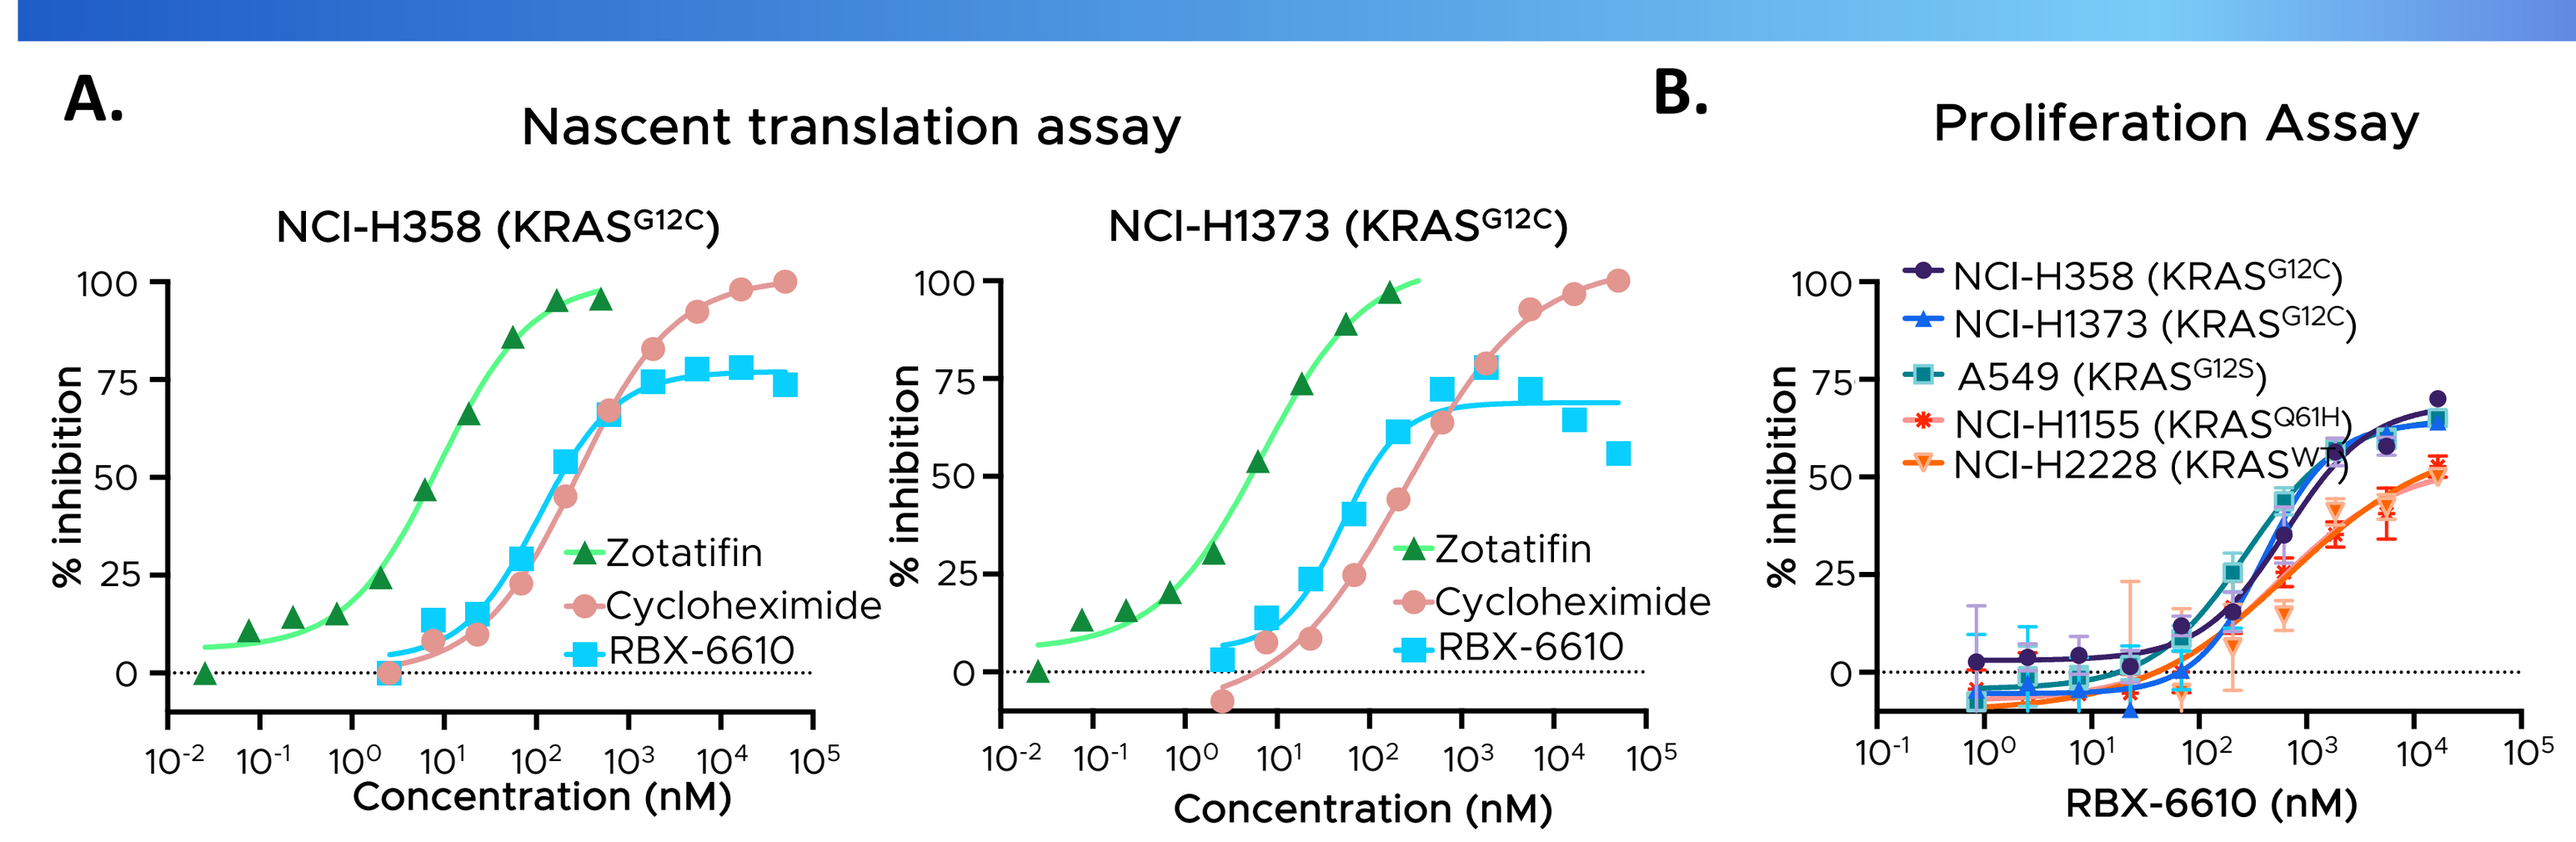
A.
Nascent translation assay
NCI-H358 (KRASG12C)
NCI-H1373 (KRASG12C)
% inhibition
Zotatifin
Cycloheximide
RBX-6610
Concentration (nM)
B.
Proliferation Assay
NCI-H358 (KRASG12C)
NCI-H1373 (KRASG12C)
A549 (KRASG12S)
NCI-H1155 (KRASQ61H)
NCI-H2228 (KRASWT)
RBX-6610 (nM)

C.

IC50 value table in proliferation assay

	IC$_{50}$ (nM)
NCI-H358	637
NCI-H1373	400
A549	275
NCI-H2228	683
NCI-H1155	541

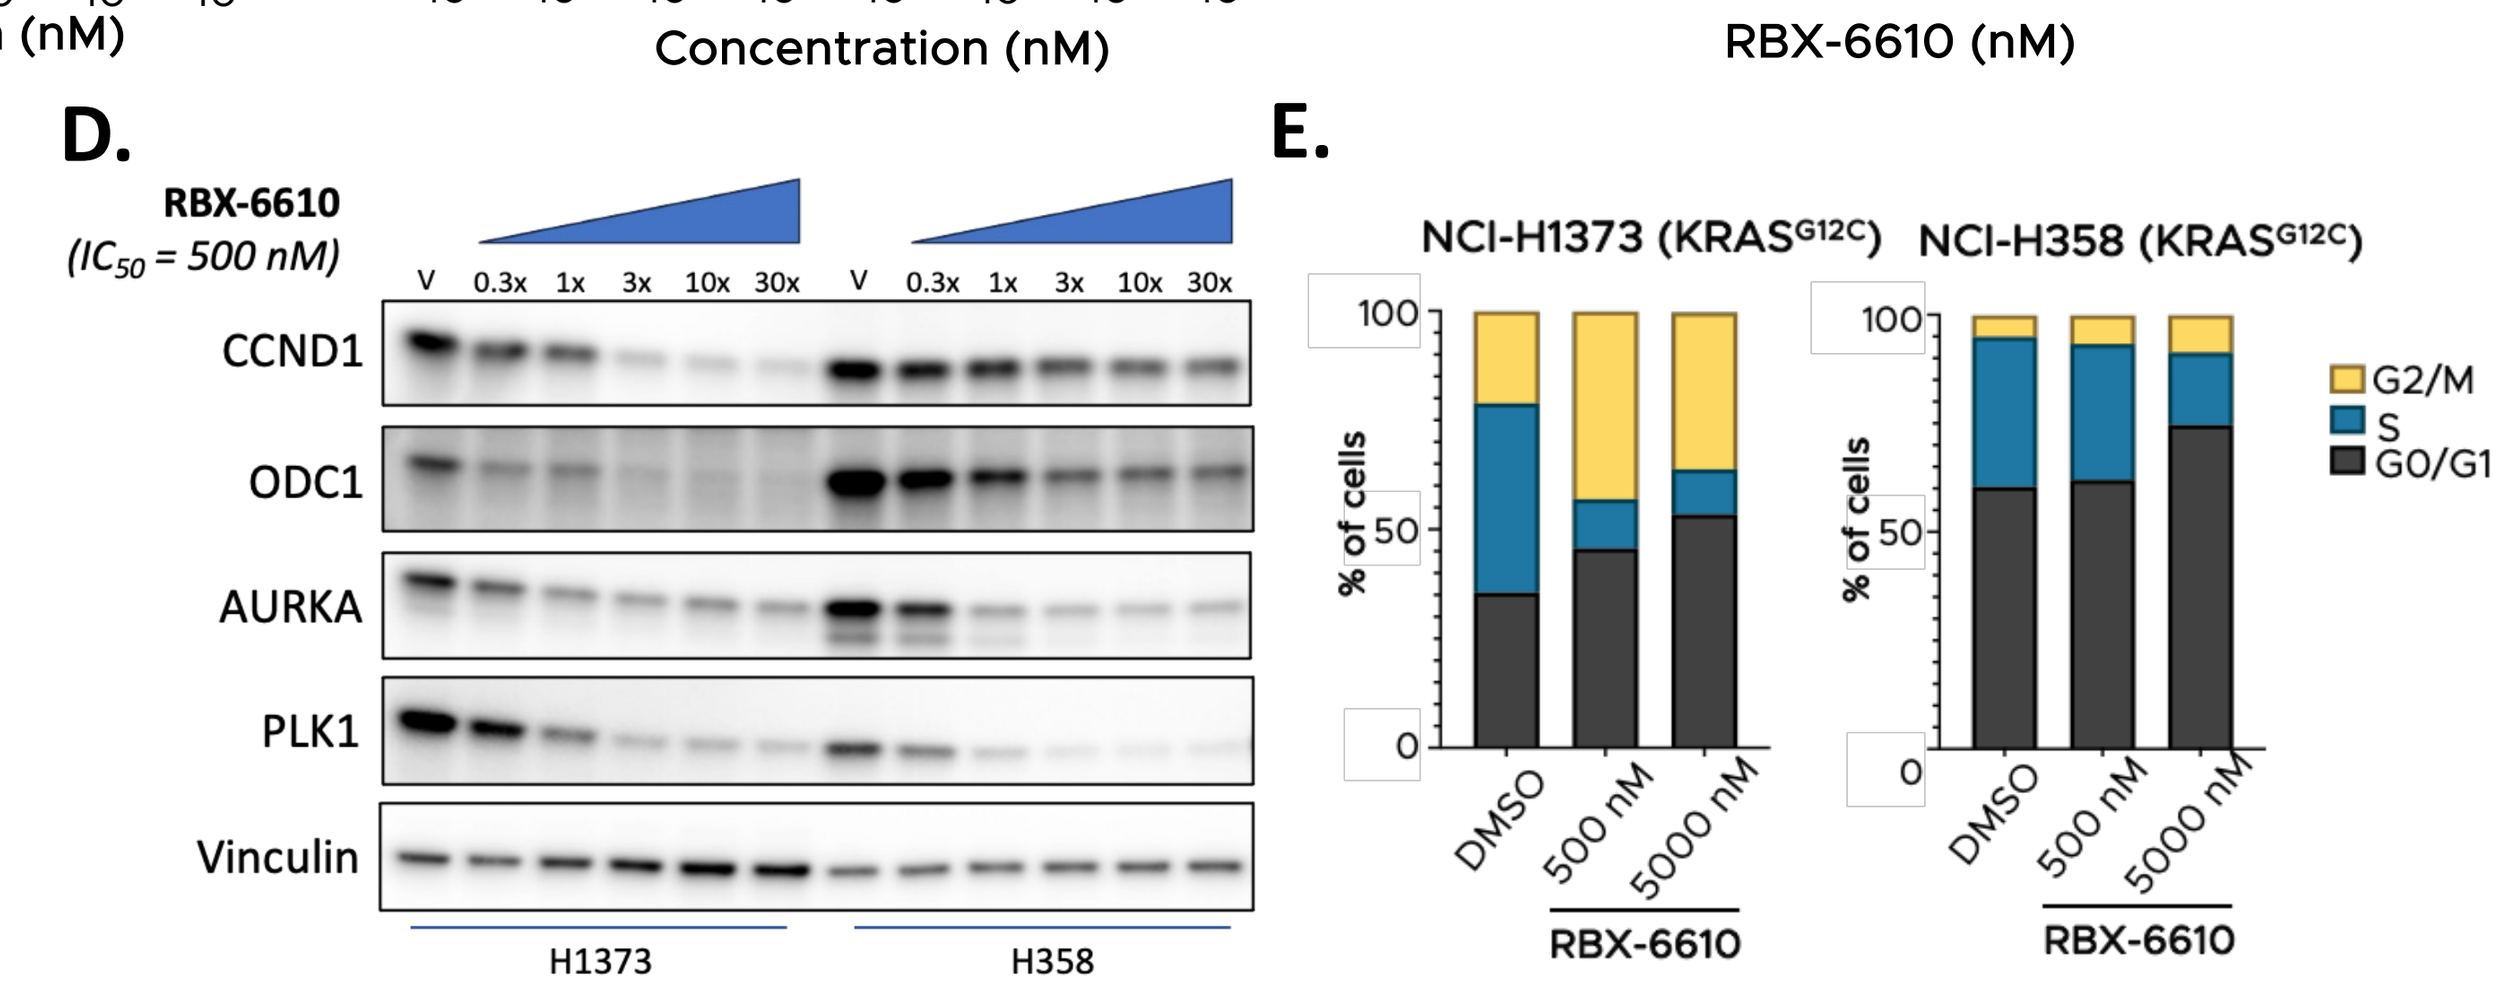
D.
RBX-6610
(IC50 = 500 nM)
V 0.3x 1x 3x 10x 30x
CCND1
ODC1
AURKA
PLK1
Vinculin
H1373 (KRASG12C)
H358 (KRASG12C)
E.
NCI-H1373 (KRASG12C)
NCI-H358 (KRASG12C)
% of cells
G2/M
S
G0/G1
DMSO
500 nM
5000 nM
RBX-6610

Figure 1. RBX-6610 exhibits inhibitory effects on translation and cancer cell proliferation. **A)** Nascent strand translation assay with O-propargyl-puromycin (OPP) demonstrates RBX-6610's mechanism of action by blocking translation. Cycloheximide is used as a positive control in this assay. Zotatifin: eIF4A inhibitor. **B)** Cell proliferation assays using CTG demonstrate RBX-6610's growth inhibitory effects across different NSCLC cell lines. **C)** Table summary of IC$_{50}$ values of RBX-6610 across the NSCLC cell lines in cell proliferation assay. **D)** Western blots of RBX-6610 treatment in NCI-H1373 and NCI-H358 KRAS^{G12C} NSCLC cell lines. **E)** Cell cycle analysis by flow cytometry in NCI-H1373 and NCI-H358 cells treated with RBX-6610.

Combination of RBX-6610 and KRAS^{G12C} inhibition induces apoptosis *in vitro*

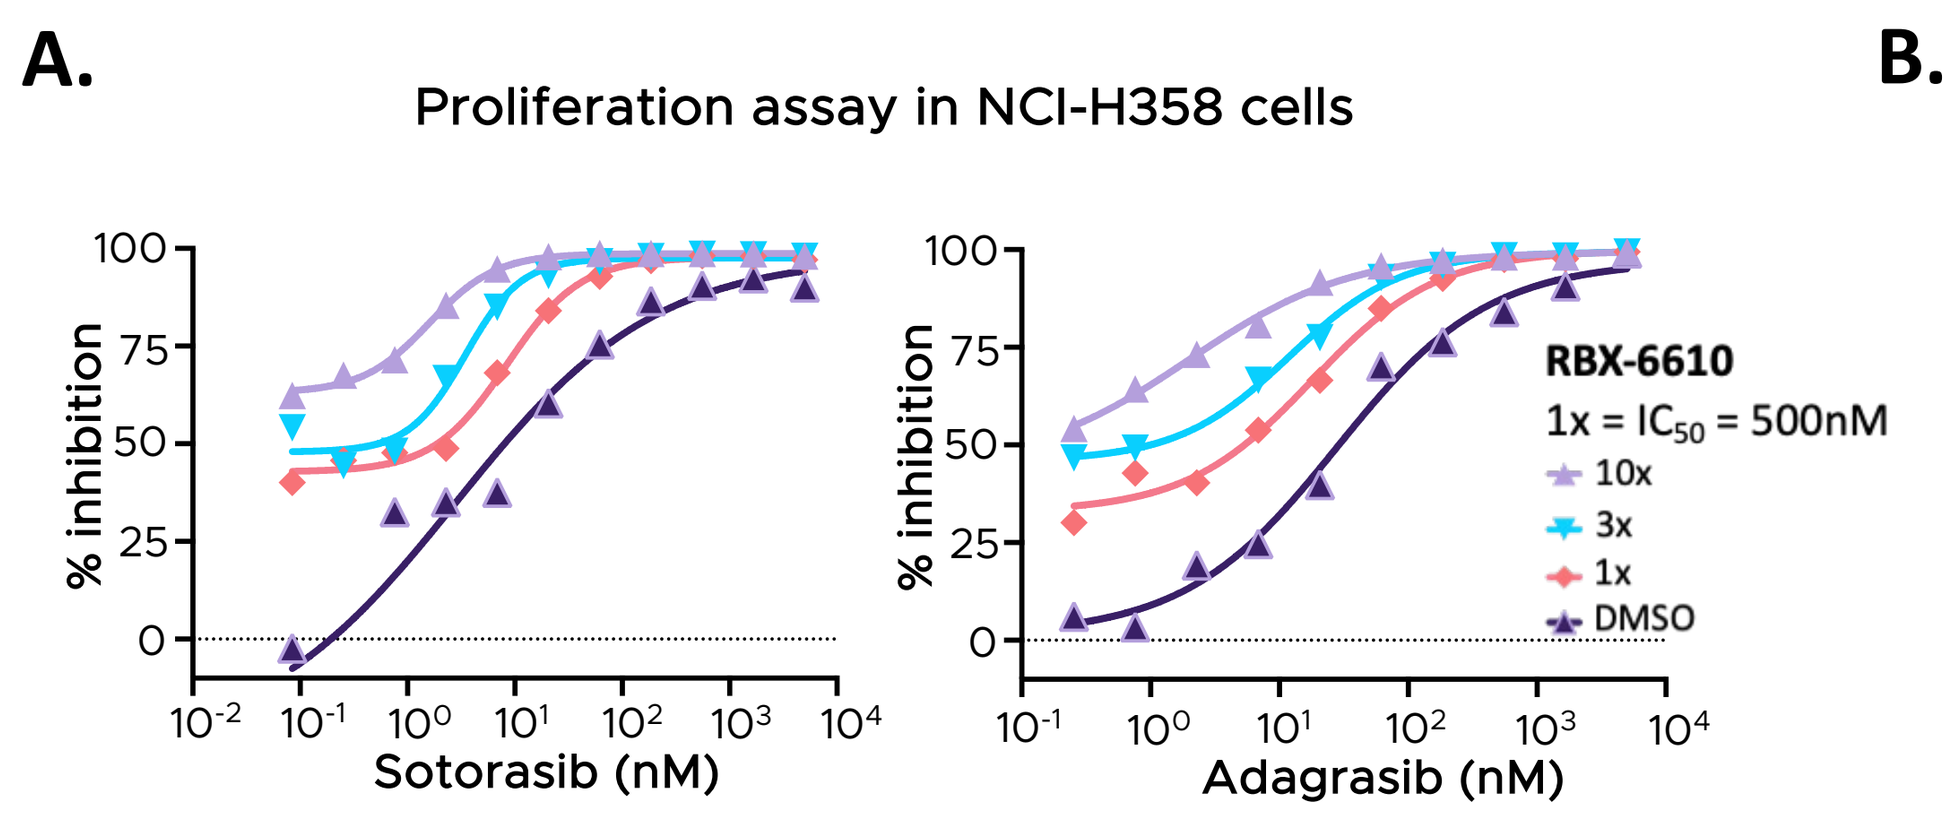
A.
Proliferation assay in NCI-H358 cells
% inhibition
Sotorasib (nM)
Adagrasib (nM)
RBX-6610
1x = IC50 = 500nM
10x
3x
1x
DMSO

RBX-6610	Sotorasib IC$_{50}$ (nM)	Adagrasib IC$_{50}$ (nM)
DMSO	24.9	27.7
1x	8.4	17.8
3x	3.5	12.0
10x	1.6	2.0

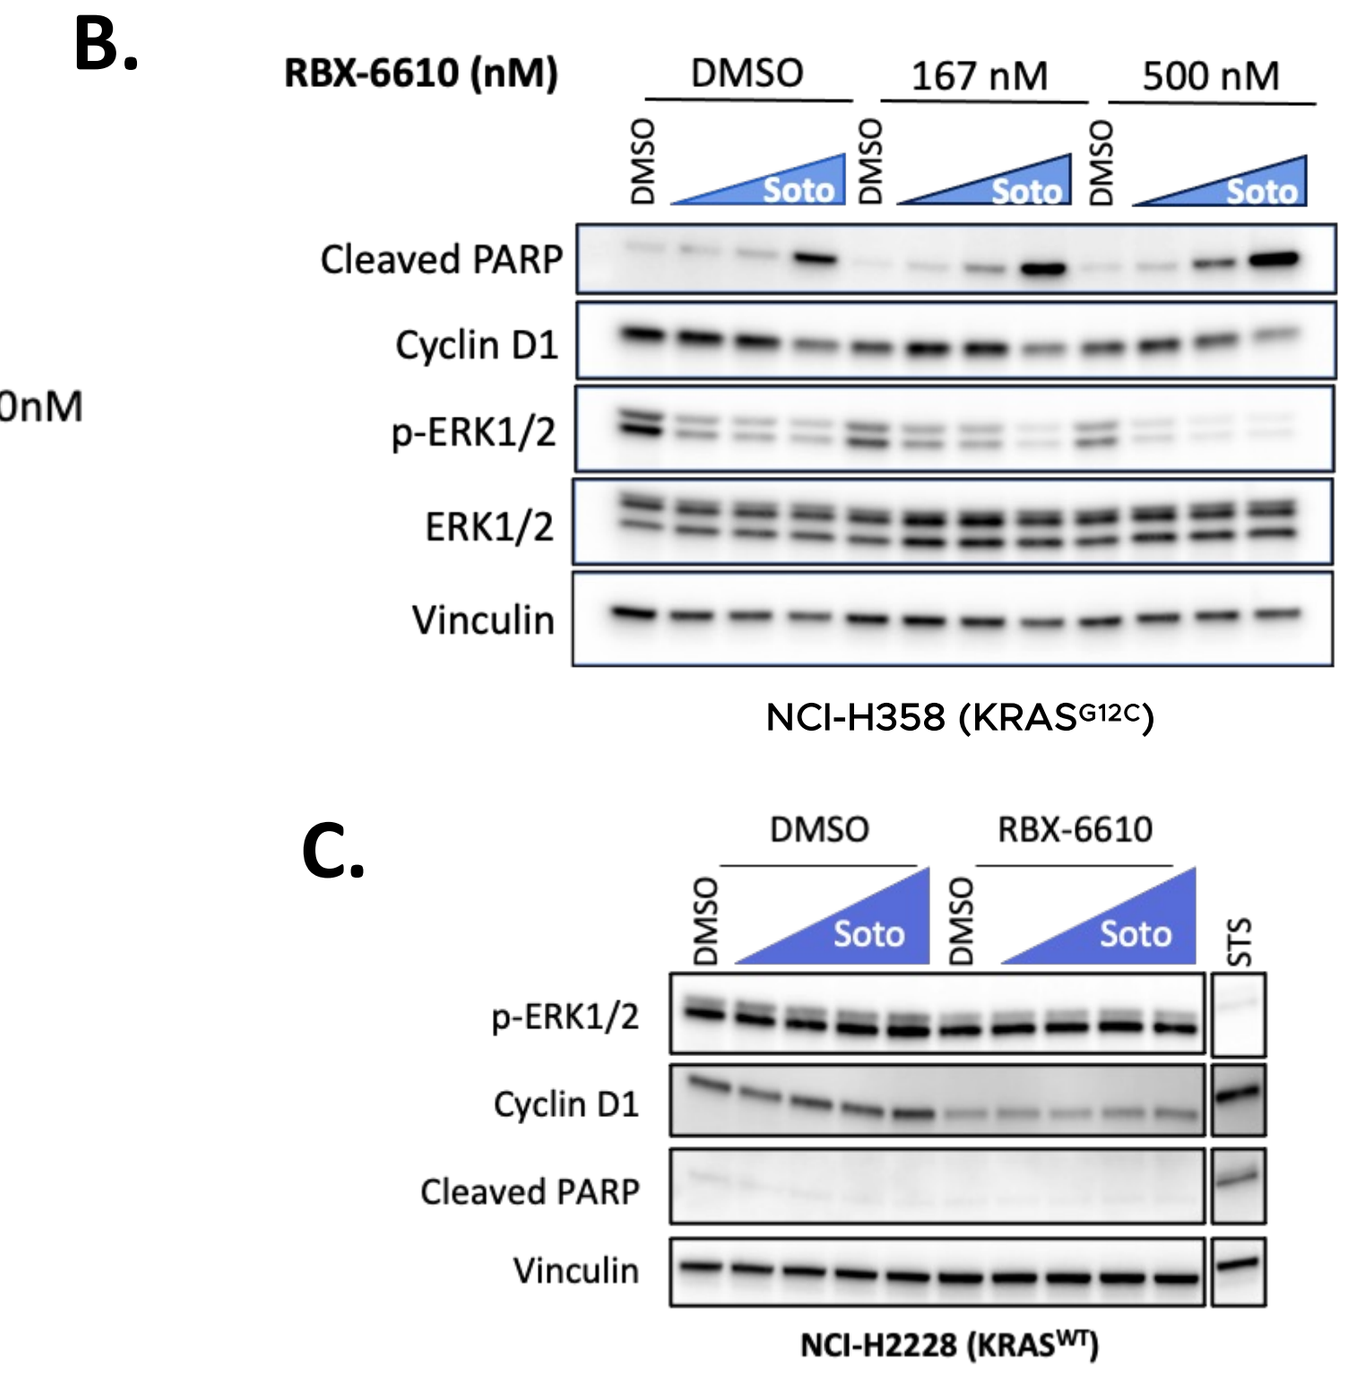
B.
RBX-6610 (nM)
DMSO
167 nM
500 nM
Soto
Cleaved PARP
Cyclin D1
p-ERK1/2
ERK1/2
Vinculin
NCI-H358 (KRASG12C)
C.
DMSO
RBX-6610
Soto
STS
p-ERK1/2
Cyclin D1
Cleaved PARP
Vinculin
NCI-H2228 (KRASWT)

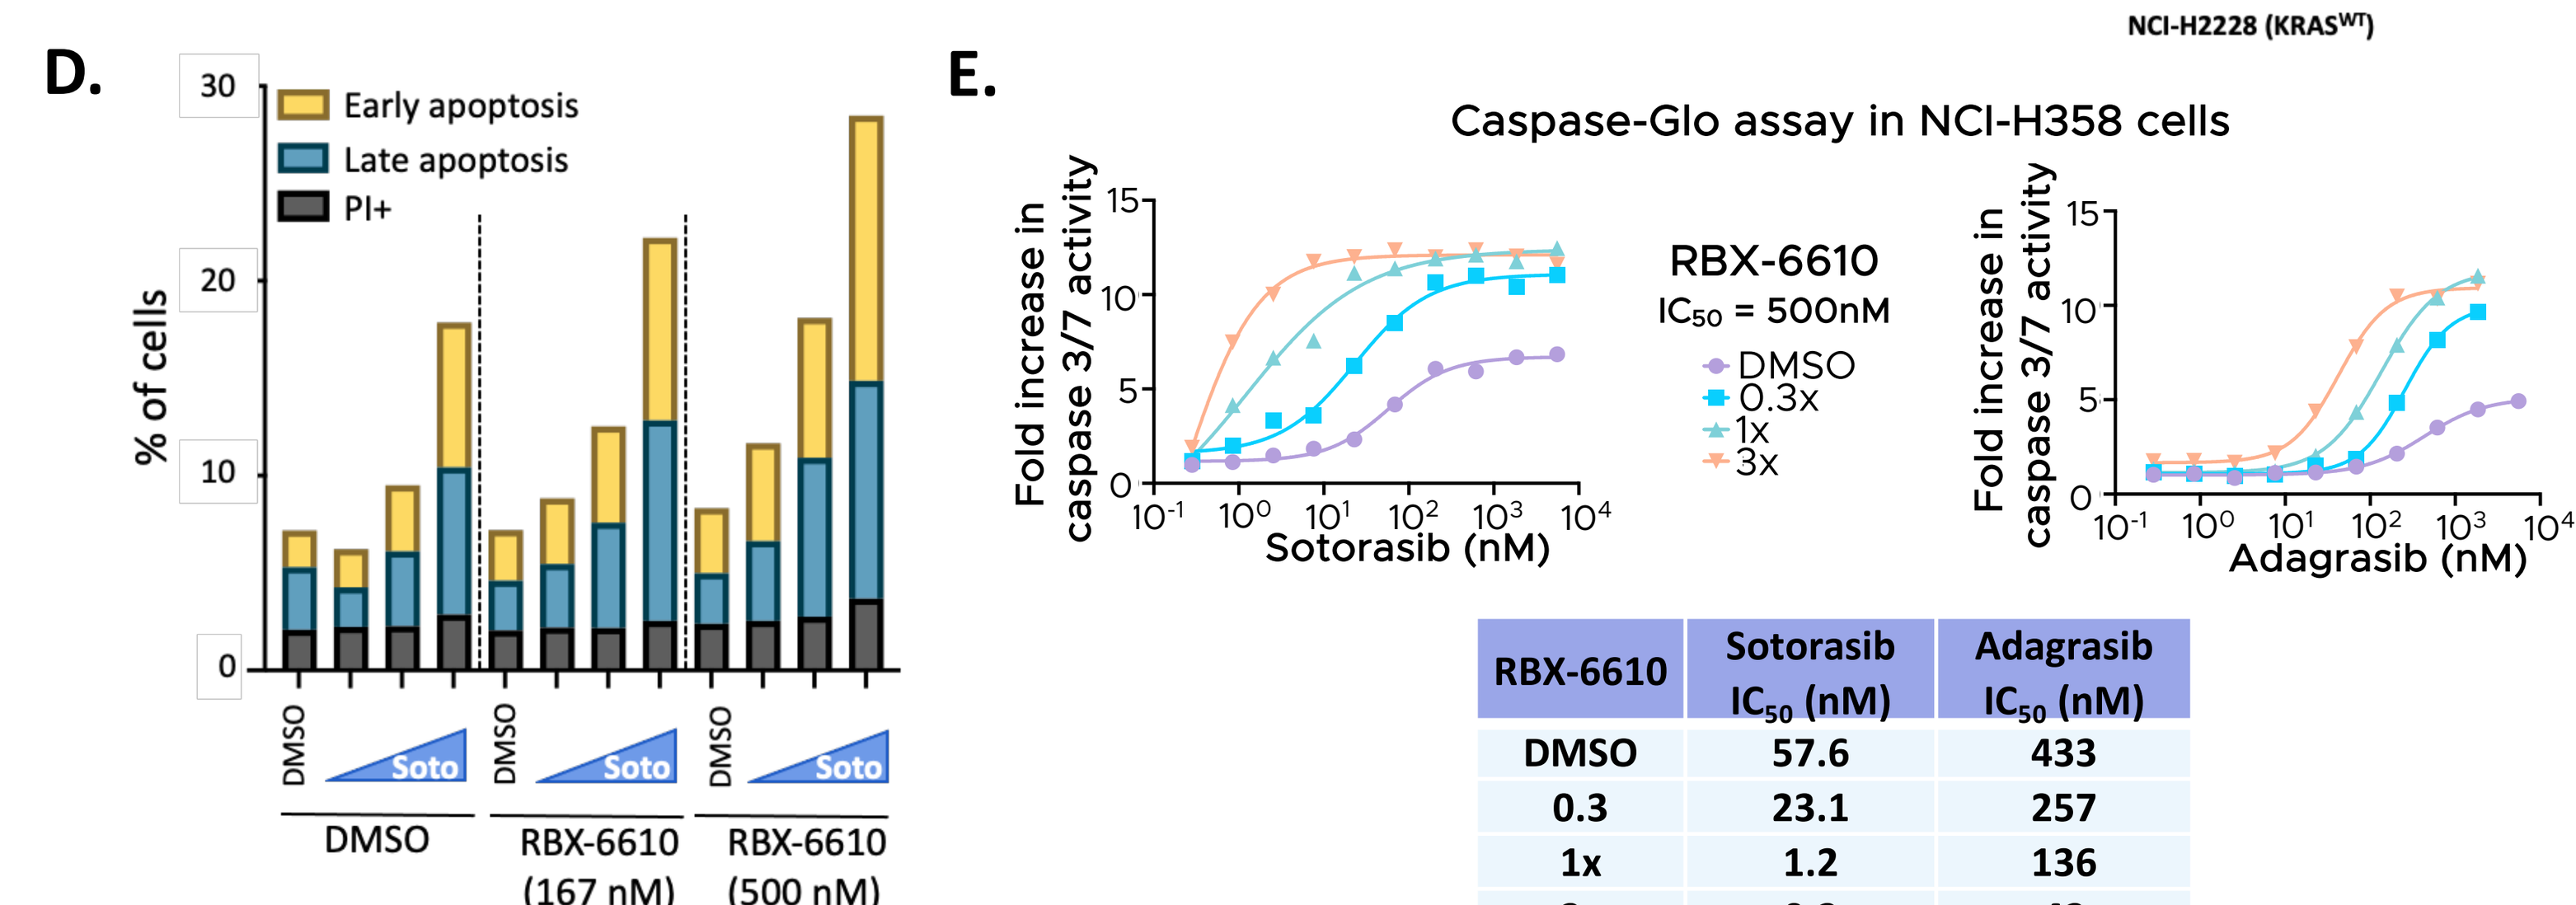
D.
Early apoptosis
Late apoptosis
PI+
% of cells
DMSO
Soto
RBX-6610 (167 nM)
RBX-6610 (500 nM)
E.
Caspase-Glo assay in NCI-H358 cells
Fold increase in caspase 3/7 activity
Sotorasib (nM)
Adagrasib (nM)
RBX-6610
IC50 = 500nM
DMSO
0.3x
1x
3x

RBX-6610	Sotorasib IC$_{50}$ (nM)	Adagrasib IC$_{50}$ (nM)
DMSO	57.6	433
0.3	23.1	257
1x	1.2	136
3x	0.2	42

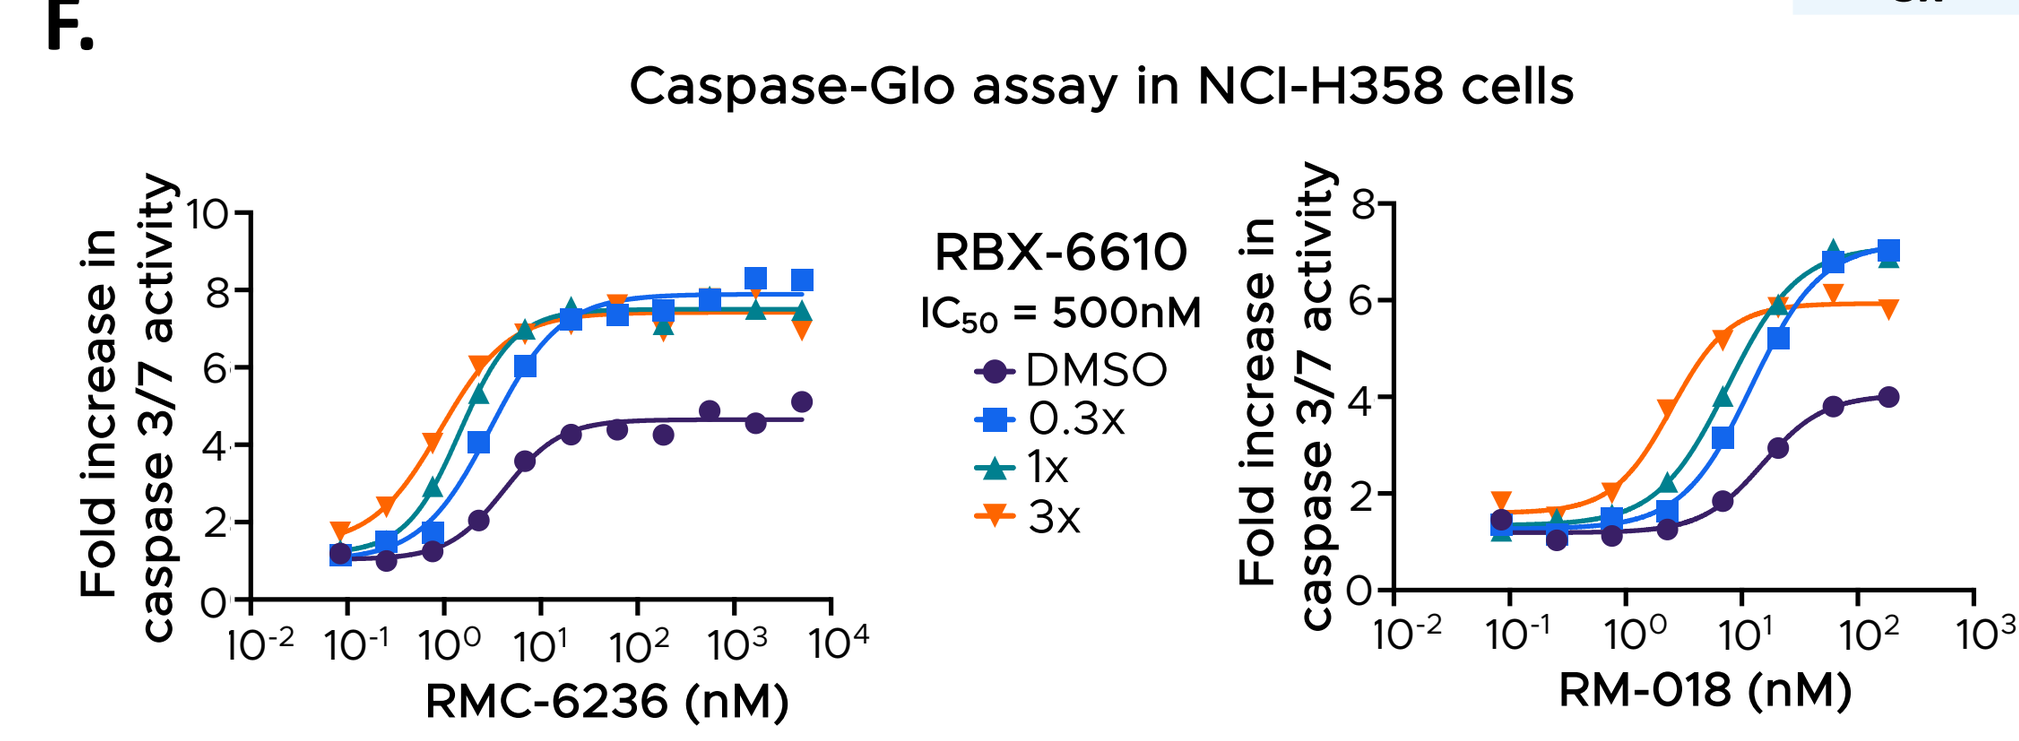
F.
Caspase-Glo assay in NCI-H358 cells
Fold increase in caspase 3/7 activity
RMC-6236 (nM)
RM-018 (nM)
RBX-6610
IC50 = 500nM
DMSO
0.3x
1x
3x

RBX-6610	RMC-6236 IC$_{50}$ (nM)	RM-018 IC$_{50}$ (nM)
DMSO	4.2	15.2
0.3	3.0	12.2
1x	1.5	7.6
3x	0.9	2.5

Figure 2. RBX-6610 synergizes with KRAS inhibitors to enhance apoptotic cell death. **A)** Cell proliferation assay using CTG demonstrates the enhanced growth inhibition effect when combining RBX-6610 with KRAS^{G12C} inhibitors in NCI-H358 cells. **B)** Western blots of combination treatment with sotorasib (Soto) and RBX-6610 in NCI-H358 cells. **C)** Western blots of combination treatment with sotorasib (Soto) and RBX-6610 in KRAS WT cells, NCI-H2228. Staurosporine (STS) used as positive control for cleaved PARP. **D)** Apoptosis flow cytometry analysis of combination treatment with sotorasib (Soto) and RBX-6610 in NCI-H358 cells. **E)** Caspase-Glo assay with KRAS^{G12C} inhibitors and RBX-6610 in NCI-H358 cells. **F)** Caspase-Glo assay with KRAS inhibitors (RM-018 and RMC-6236) and RBX-6610 in NCI-H358 cells. RM-018: KRAS^{G12C} (ON) inhibitor. RMX-6236: Pan RAS (ON) inhibitor.

RBX-6610 synergizes with sotorasib in treatment naïve KRAS^{G12C} NSCLC *in vivo*

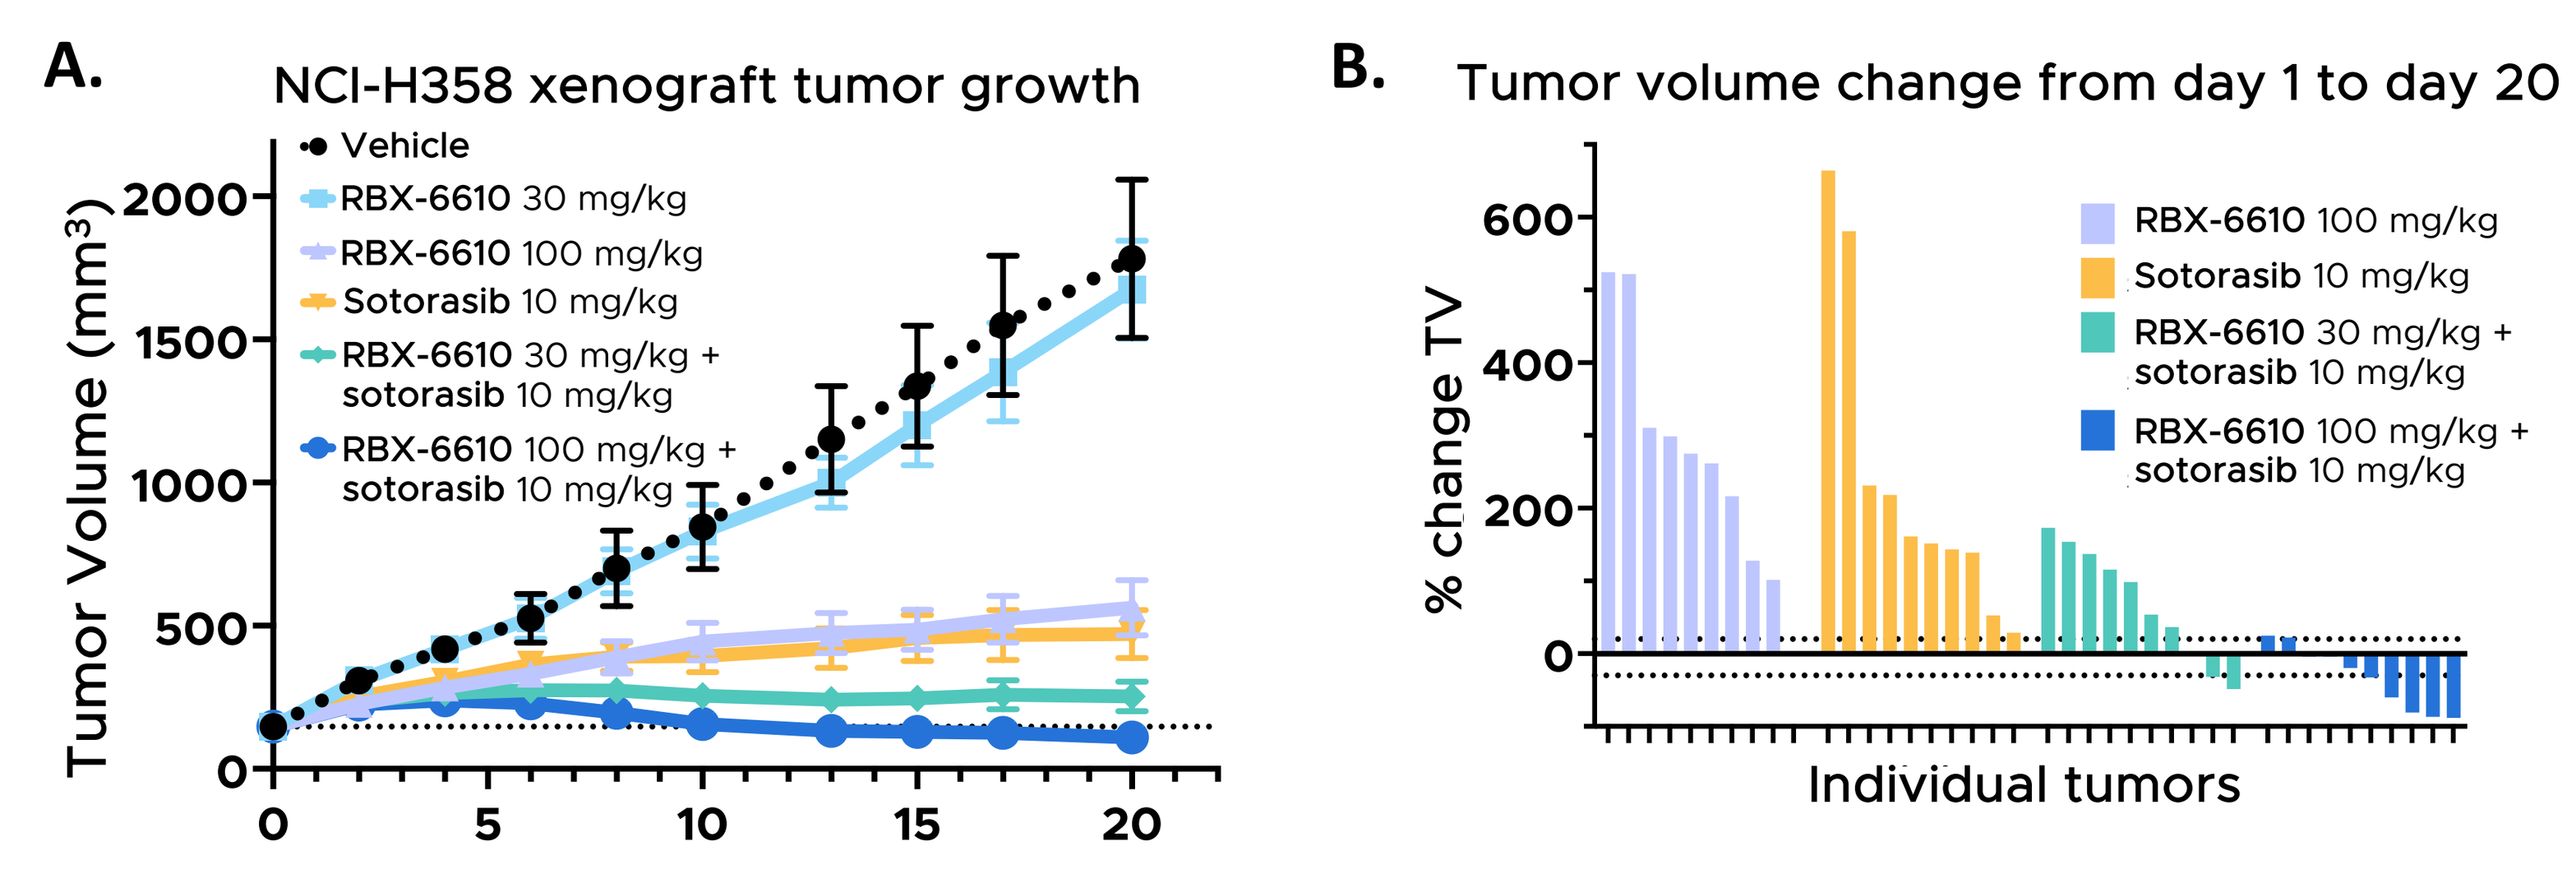
A.
NCI-H358 xenograft tumor growth
Vehicle
RBX-6610 30 mg/kg
RBX-6610 100 mg/kg
Sotorasib 10 mg/kg
RBX-6610 30 mg/kg + sotorasib 10 mg/kg
RBX-6610 100 mg/kg + sotorasib 10 mg/kg
Tumor Volume (mm3)
mean TV ± SEM
Days
B.
Tumor volume change from day 1 to day 20
% change TV
Individual tumors

C.

Synergy combination index scores

	Bliss CI†	HSA CI†
RBX-6610 30 mg/kg + sotorasib 10 mg/kg	0.23	0.27
RBX-6610 100 mg/kg + sotorasib 10 mg/kg	0.13	0.63

All groups dosed QD x20 days, p.o.; female NCr nu/nu mice, n=10 per group

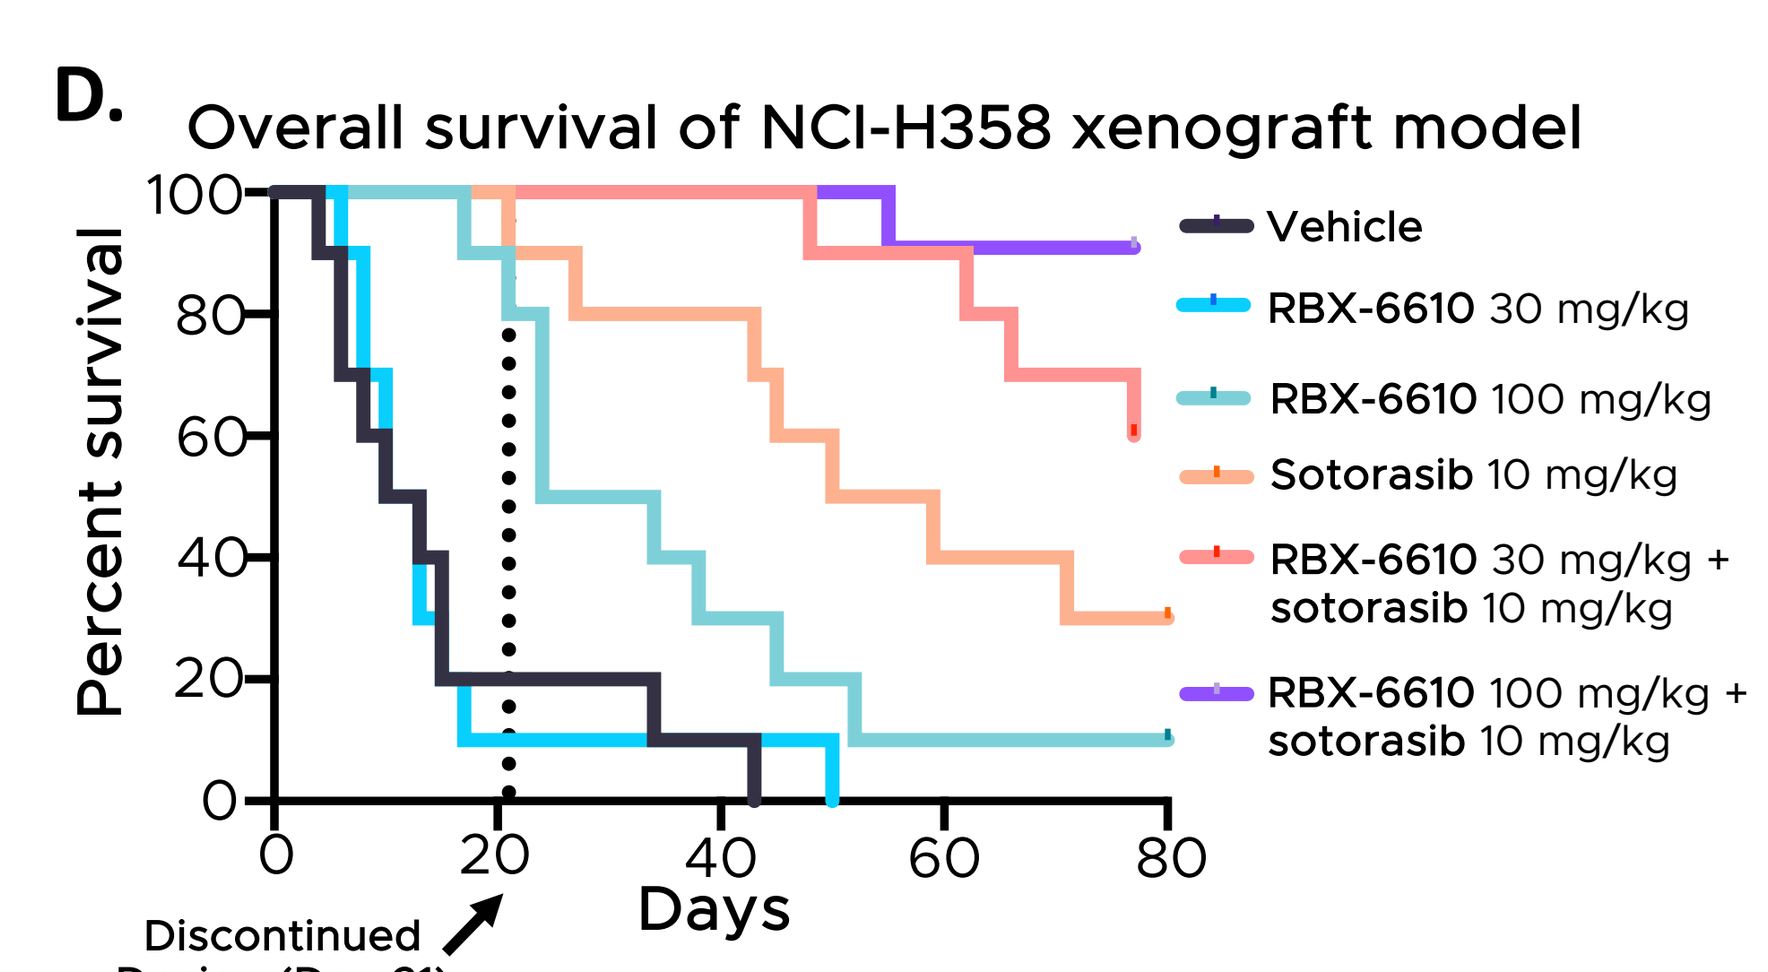
D.
Overall survival of NCI-H358 xenograft model
Percent survival
Days
Discontinued Dosing (Day 21)
Vehicle
RBX-6610 30 mg/kg
RBX-6610 100 mg/kg
Sotorasib 10 mg/kg
RBX-6610 30 mg/kg + sotorasib 10 mg/kg
RBX-6610 100 mg/kg + sotorasib 10 mg/kg

Figure 3. RBX-6610 synergizes with KRAS inhibitors to cause tumor regression in a KRASG12C NSCLC model. **A)** NCI-H358 xenograft tumor growth curves with daily oral treatment of sotorasib, RBX-6610, and RBX-6610 plus sotorasib. **B)** Percent change in tumor volume for individual mice. Dashed lines indicate cutoffs for: progressive disease >+20%, stable disease <+20% and >-30%, and partial response <-30%. **C)** Synergy combination index scores for combination of sotorasib with two RBX-6610 concentrations. Bliss and HSA (highest single agent) combination index scores where >0 is synergistic, =0 is additive, and <0 is antagonistic. Method from Huang et al., *Sci Rep* **12**, 12984 (2022). **D)** Overall survival curves of NCI-H358 xenograft mice. Vertical dashed line indicates when the last treatments were administered.

RBX-6610 demonstrates *in vitro* activity in *KRASi* resistant cell lines

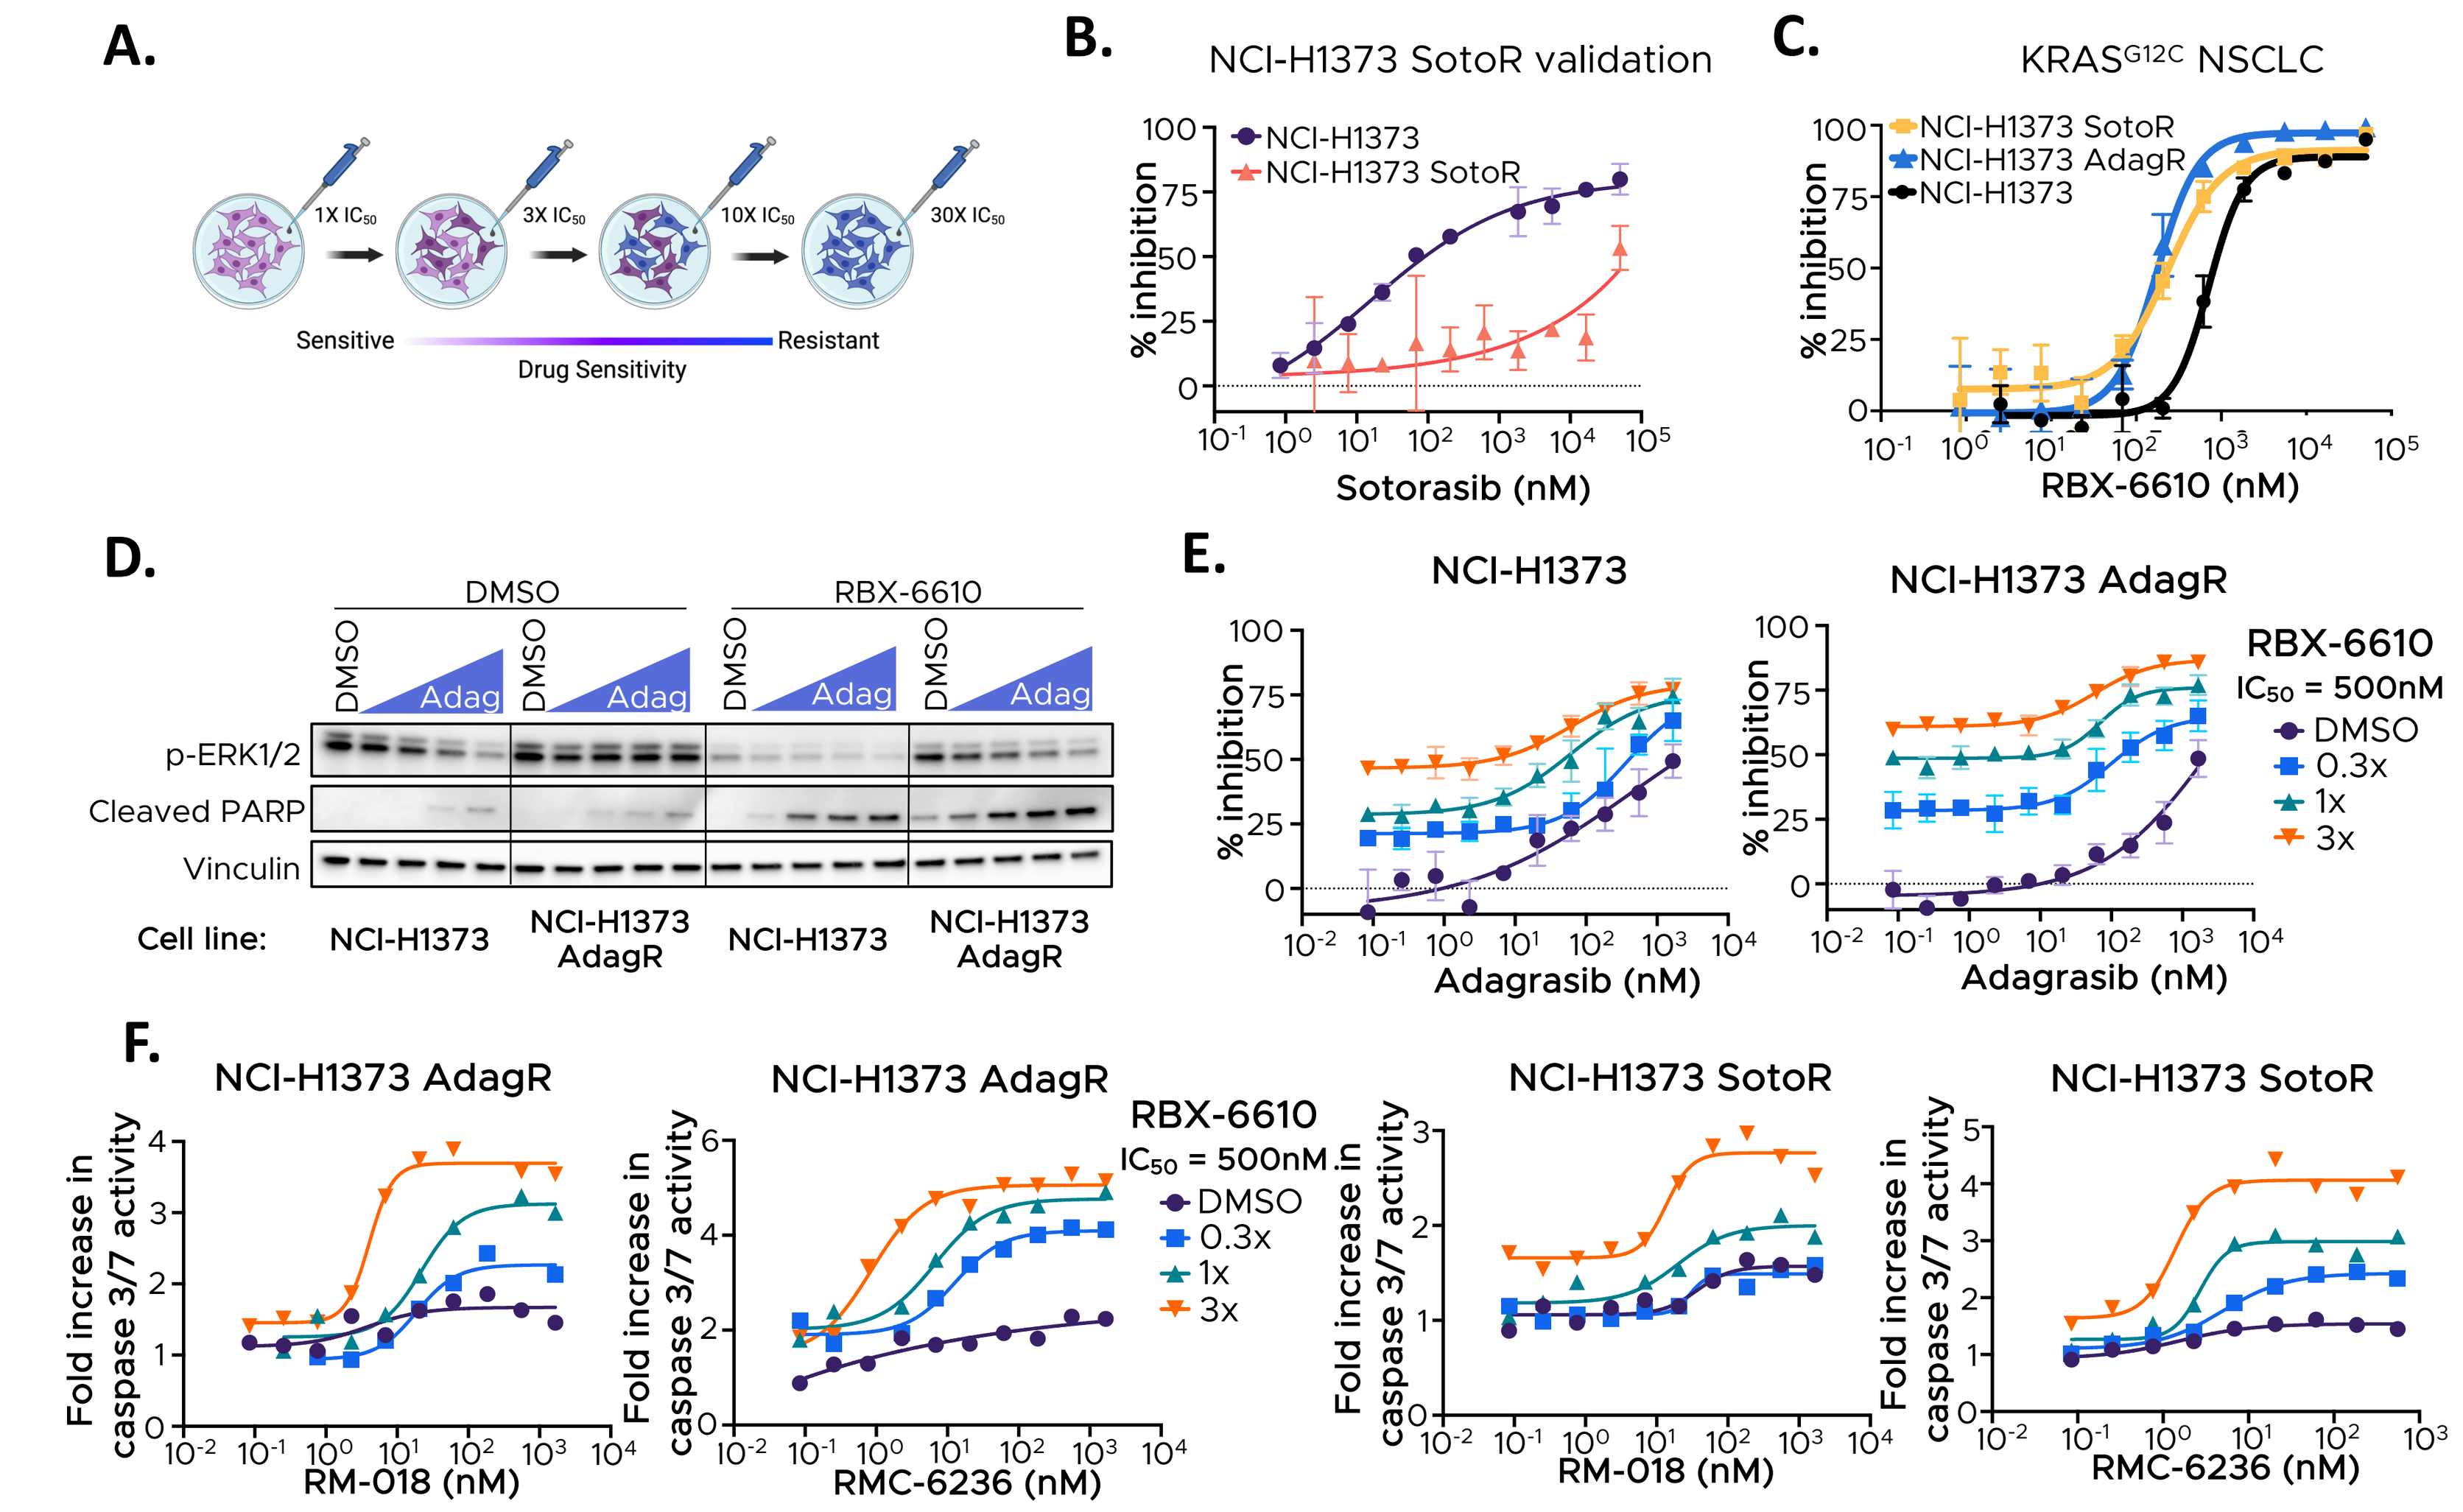
A.
1X IC50
3X IC50
10X IC50
30X IC50
Sensitive
Drug Sensitivity
Resistant
B.
NCI-H1373 SotoR validation
NCI-H1373
NCI-H1373 SotoR
% inhibition
Sotorasib (nM)
C.
KRASG12C NSCLC
NCI-H1373 SotoR
NCI-H1373 AdagR
NCI-H1373
RBX-6610 (nM)
D.
DMSO
RBX-6610
Adag
p-ERK1/2
Cleaved PARP
Vinculin
Cell line:
NCI-H1373
NCI-H1373 AdagR
E.
NCI-H1373
NCI-H1373 AdagR
Adagrasib (nM)
RBX-6610
IC50 = 500nM
DMSO
0.3x
1x
3x
F.
NCI-H1373 AdagR
NCI-H1373 SotoR
Fold increase in caspase 3/7 activity
RM-018 (nM)
RMC-6236 (nM)

Figure 4. Sotorasib and adagrisib resistant cancer cells are more dependent on eIF4E for growth and RBX-6610 re-sensitizes resistant lines to SOC treatment. **A)** Serial passaging strategy to generate acquired resistance cell lines. **B)** Validation of sotorasib resistance in NCI-H1373 SotoR cells using CTG assay. **C)** Anti-proliferative effects of RBX-6610 on sotorasib and adagrasib resistant NCI-H1373 cells. **D)** Western blots of combination treatment with adagrasib (Adag) and RBX-6610 in parental and resistant cells. **E)** Growth inhibition assays using CTG demonstrate re-sensitization of adagrasib NCI-H1373 cells to the KRAS^{G12C} inhibitors. **F)** Caspase-Glo assay with KRAS inhibitors, RM-018 and RMC-6236, in sotorasib and adagrasib resistant NCI-H1373 cells.

RBX-6610 re-sensitizes sotorasib in *KRASi* resistant KRAS^{G12C} NSCLC

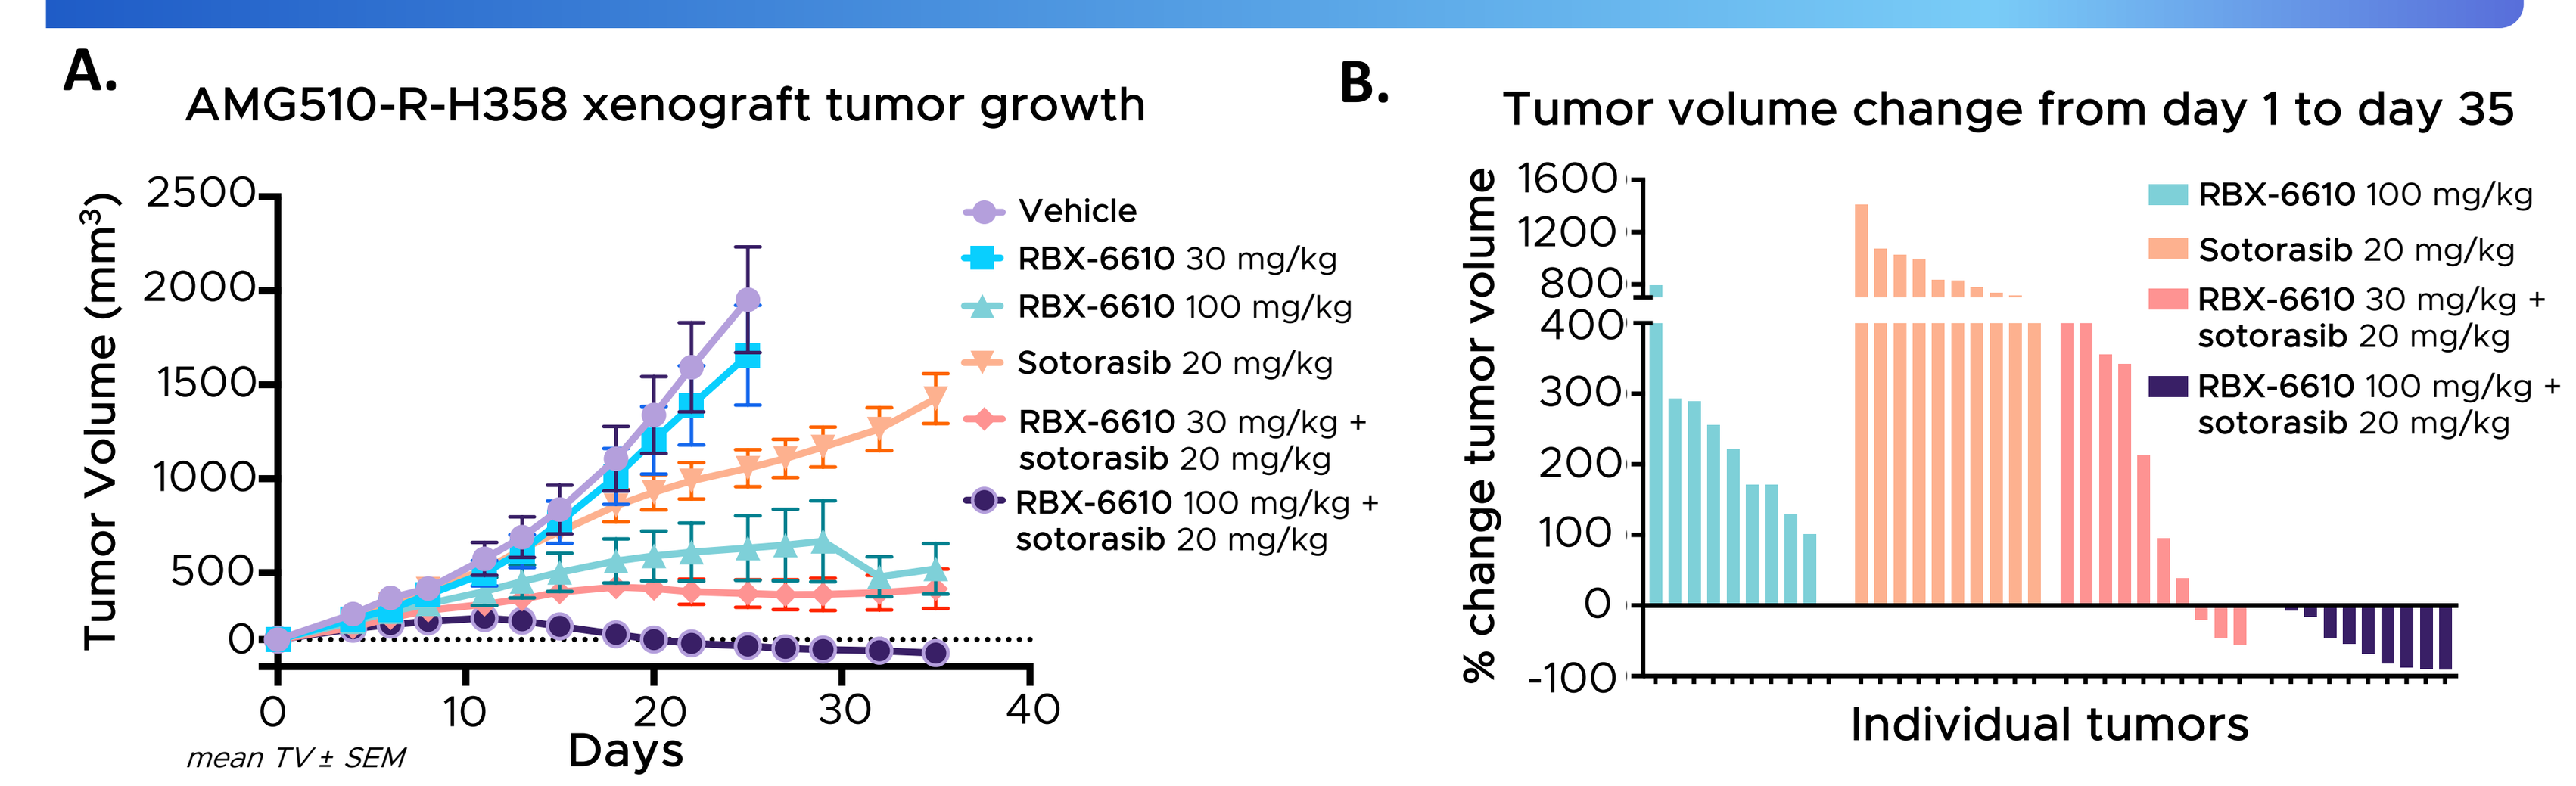
A.
AMG510-R-H358 xenograft tumor growth
Vehicle
RBX-6610 30 mg/kg
RBX-6610 100 mg/kg
Sotorasib 20 mg/kg
RBX-6610 30 mg/kg + sotorasib 20 mg/kg
RBX-6610 100 mg/kg + sotorasib 20 mg/kg
Tumor Volume (mm3)
mean TV ± SEM
Days
B.
Tumor volume change from day 1 to day 35
% change tumor volume
Individual tumors

C.

Summary of treatment outcomes

	PD	SD	PR	CR
Vehicle	10	0	0	0
RBX-6610, 30 mpk	10	0	0	0
RBX-6610, 100 mpk	9	1	0	0
Sotorasib, 20 mpk	10	0	0	0
RBX, 30 mpk + soto	7	1	2	0
RBX, 100 mpk + soto	0	3	3	4

PD (progressive disease) >30%
SD (stable disease) <30% & >-30%
PR (partial response) < -30% & >-80%
CR (complete response) >-80%

D.

Treatment group	%TGI† Naïve H358	%TGI† Resistant H358	Mean % relative change TV Naïve H358 (d21)	Mean % relative change TV Resistant H358 (d35)
Vehicle	n/a	n/a	+1170%	+1251%
RBX-6610, 30 mpk	7%	10%	+998%	+1047%
RBX-6610, 100 mpk	75%	71%	+288%	+260%
Sotorasib, 10 mpk (naïve) Sotorasib, 20 mpk (resist)	80%	44%	+250%	+887%
RBX, 30 mpk + sotorasib	94%	86%	+71%	+186%
RBX, 100 mpk + sotorasib	102%	103%	-34%	-49%

†Tumor growth inhibition; % change in tumor growth relative to vehicle tumor growth, excluding ulcerated tumors.

Figure 5. RBX-6610 demonstrates anti-tumor effects in sotorasib resistant NCI-H358 xenograft model (AMG510-R-H358) and re-sensitizes tumors to sotorasib. **A)** AMG-510-R-H358 xenograft tumor growth curves with daily oral treatment of sotorasib, RBX-6610, and RBX-6610 plus sotorasib. **B)** Percent change in tumor volume for individual mice in the indicated treatment groups. **C)** Table summarizes the number of mice with progressive disease, stable disease, partial response and complete response in each treatment group. **D)** Summary table compares the anti-tumor efficacy of the indicated treatments between naïve (Figure 3) and sotorasib resistant NCI-H358 xenograft models.

Conclusions

- Daily oral dosing of RBX-6610 shows *in vivo* anti-tumor efficacy in KRAS^{G12C} mutant NSCLC xenografts
- RBX-6610 synergistically combines with KRAS^{G12C} inhibitors for enhanced *in vivo* anti-tumor efficacy in both the treatment naïve and treatment resistant setting with no observed toxicity after up to 35 days of dosing in mice
- *In vitro*, the combination of RBX-6610 with KRAS inhibitors (including approved KRAS^{G12C} inhibitors, and investigational RAS inhibitors) lead to an apoptotic phenotype supporting the significant efficacy observed in the *in vivo* studies
- Non GLP studies have been completed; IND-enabling studies are ongoing

ESMO Annual Meeting 2024 , Sept. 13th-17th 2024, Barcelona

Contact email address: jsorrentino@ribometrix.com