Leveraging an RNA-targeting platform for the discovery of cell-active c-MYC mRNA-binding small molecules

No. 680

Matthew J Smola, Krista Marran, Sarah EG Thompson, Brittani Patterson, Roheeth Kumar Pavana, Caleb Sutherland, Raquel C Martínez-Chacín, Jessica A Sorrentino, Katherine Deigan Warner

Ribometrix, Inc. Durham, NC

ABSTRACT

The MYC gene, with its oncogenic potential, has long presented a formidable challenge to conventional drug discovery efforts, and its critical role in cancer progression and resistance has underscored the need for innovative therapeutic strategies. Here we demonstrate the capabilities of the Ribometrix RNA-targeting platform to modulate the c-MYC mRNA with small molecules with the aim of reducing c-MYC protein levels.

Using our comprehensive platform of RNA-targeting drug discovery tools and analyses including chemical probing, structure determination, high-purity RNA production, high-throughput screening, and biophysical characterization, we evaluated the potential to directly target c-MYC mRNA using small molecules. Our analysis revealed a high-confidence structured element within the c-MYC mRNA 5' UTR expected to harbor a tertiary structure amenable to drug-like compound binding. Leveraging the multifaceted chemical probing and structure modeling components of our platform, we confirmed that our *in vitro*-transcribed RNA screening construct adopts the same structures found in endogenous cellular c-MYC transcripts. After large-scale *in vitro* RNA preparation, we conducted high-throughput screening of a chemically diverse drug-like library using a mass spectrometry automated ligand identification system (ALIS) that identified multiple chemical series with low micromolar affinity or better. To eliminate pan-binding compounds, we evaluated selectivity against a panel of non-target mRNA structures. Importantly, these compounds lead to rapid (4 hr) reduction of MYC protein levels in a small cell lung cancer cell line with high c-MYC expression, DMS-273, suggesting an on-target effect. Employing our suite of chemical probing tools, we confirmed that compounds from multiple series directly engage the c-MYC mRNA in cells. We prioritized medicinal chemistry efforts for Series 1 which are ongoing to increase compound potency and define the precise mechanism of action of c-MYC protein reduction in clinically relevant models.

The discovery of these c-MYC mRNA-binding small molecules not only validates the utility of the Ribometrix RNA-targeted small molecule discovery platform but also showcases its potential in tackling traditionally 'undruggable' targets, and provides a promising avenue for developing novel anti-cancer therapies. As we further refine our c-MYC mRNA-binding small molecules, our platform will continue to provide practical insights into the intricacies of c-MYC mRNA biology and previously-unknown MYC vulnerabilities, and present tangible opportunities for advancing targeted cancer therapies.

INTRODUCTION

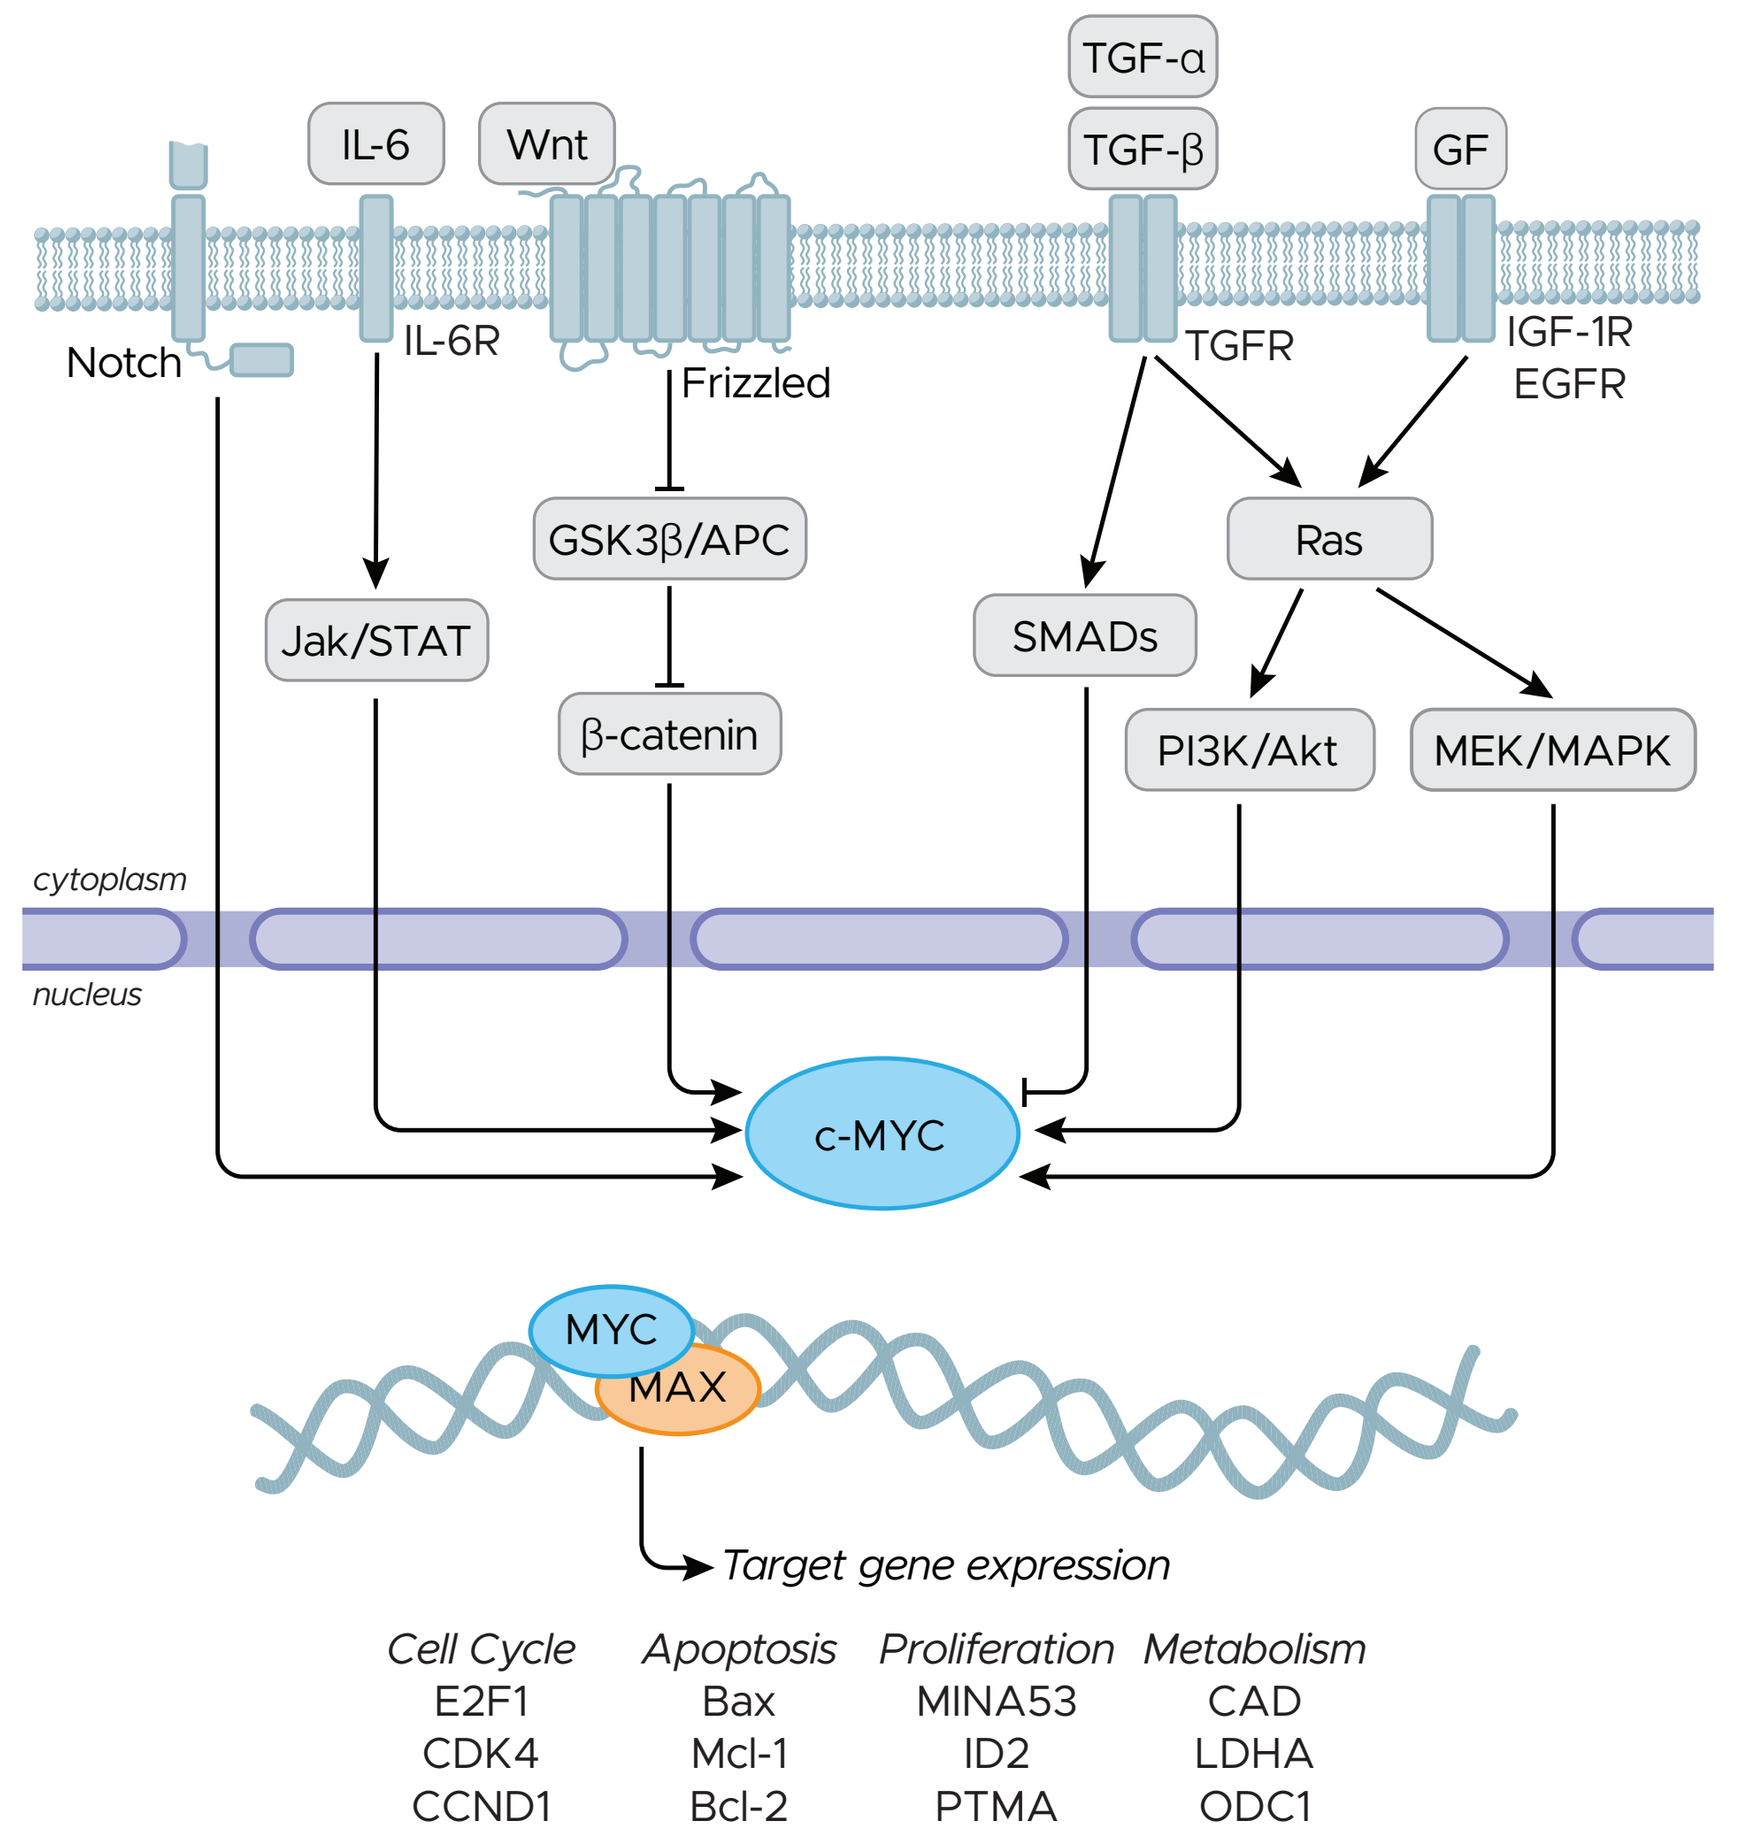
TGF-α
TGF-β
IL-6
Wnt
GF
Notch
IL-6R
Frizzled
TGFR
IGF-1R
EGFR
GSK3β/APC
Ras
Jak/STAT
SMADs
β-catenin
PI3K/Akt
MEK/MAPK
cytoplasm
nucleus
c-MYC
MYC
MAX
Target gene expression
Cell Cycle
E2F1
CDK4
CCND1
Apoptosis
Bax
Mcl-1
Bcl-2
Proliferation
MINA53
ID2
PTMA
Metabolism
CAD
LDHA
ODC1

MYC is a well-validated oncogene across many cancer indications representing a broad anti-tumor opportunity

- Dysregulated in >70% of cancers across heme and solid malignancies
- Key regulator in almost every aspect of the oncogenic process

Strong genetic and pharmacological validation

- *MYC* overexpression leads to tumorigenesis in a variety of tissues
- Tumorigenesis and hyperproliferation are significantly reduced across multiple indications in conditional *MYC* knockout mice.
- A c-MYC targeting peptide (OMO-103) showed anticancer activity in various preclinical models and is currently in clinical trials

c-MYC protein is widely deemed an "intractable" therapeutic target

- Intrinsically disordered structure and lack of defined small molecule binding site
- Small molecules that bind to c-MYC are likely to target other MYC homologs
- Inaccessibility to antibody treatment due to predominant nuclear localization

The Ribometrix platform unlocks the ability to target MYC, a validated yet intractable protein, by drugging its mRNA

METHODS

The Ribometrix MaP Suite enables RNA structure determination and compound target engagement in cells and *in vitro*

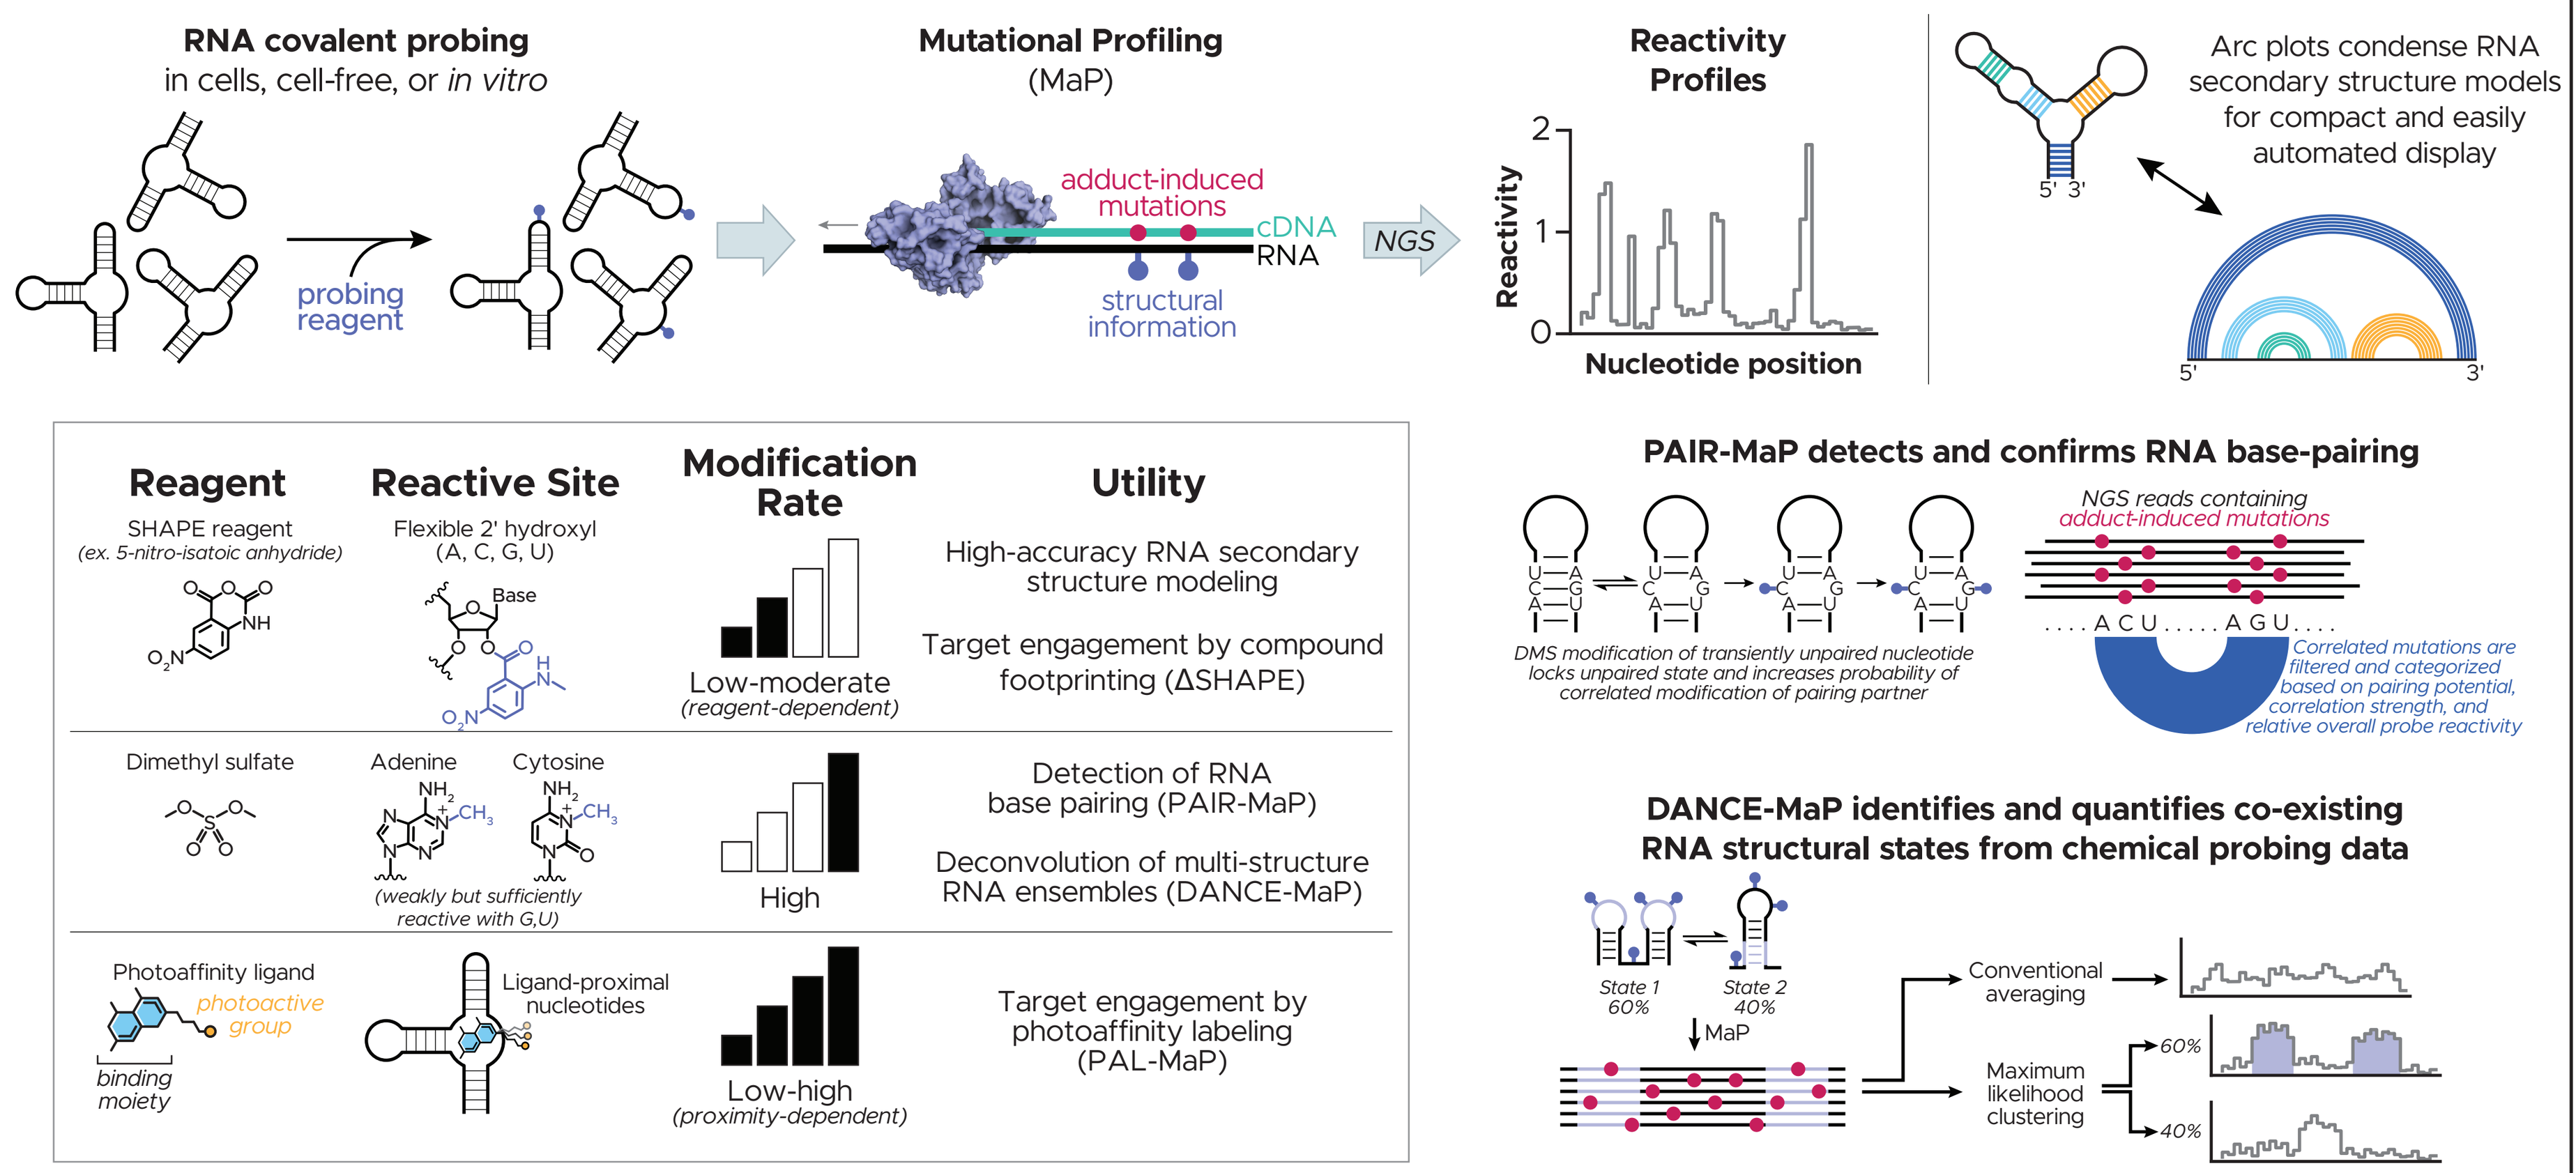
RNA covalent probing
in cells, cell-free, or in vitro
probing reagent
Mutational Profiling
(MaP)
adduct-induced mutations
cDNA
RNA
structural information
NGS
Reactivity Profiles
Reactivity
Nucleotide position
Arc plots condense RNA secondary structure models for compact and easily automated display
Reagent
Reactive Site
Modification Rate
Utility
SHAPE reagent
(ex. 5-nitro-isatoic anhydride)
Flexible 2' hydroxyl
(A, C, G, U)
Low-moderate
(reagent-dependent)
High-accuracy RNA secondary structure modeling
Target engagement by compound footprinting (ΔSHAPE)
Dimethyl sulfate
Adenine
Cytosine
(weakly but sufficiently reactive with G,U)
High
Detection of RNA base pairing (PAIR-MaP)
Deconvolution of multi-structure RNA ensembles (DANCE-MaP)
Photoaffinity ligand
photoactive group
binding moiety
Ligand-proximal nucleotides
Low-high
(proximity-dependent)
Target engagement by photoaffinity labeling (PAL-MaP)
PAIR-MaP detects and confirms RNA base-pairing
NGS reads containing adduct-induced mutations
DMS modification of transiently unpaired nucleotide locks unpaired state and increases probability of correlated modification of pairing partner
Correlated mutations are filtered and categorized based on pairing potential, correlation strength, and relative overall probe reactivity
DANCE-MaP identifies and quantifies co-existing RNA structural states from chemical probing data
State 1
60%
State 2
40%
MaP
Conventional averaging
Maximum likelihood clustering
60%
40%

RESULTS

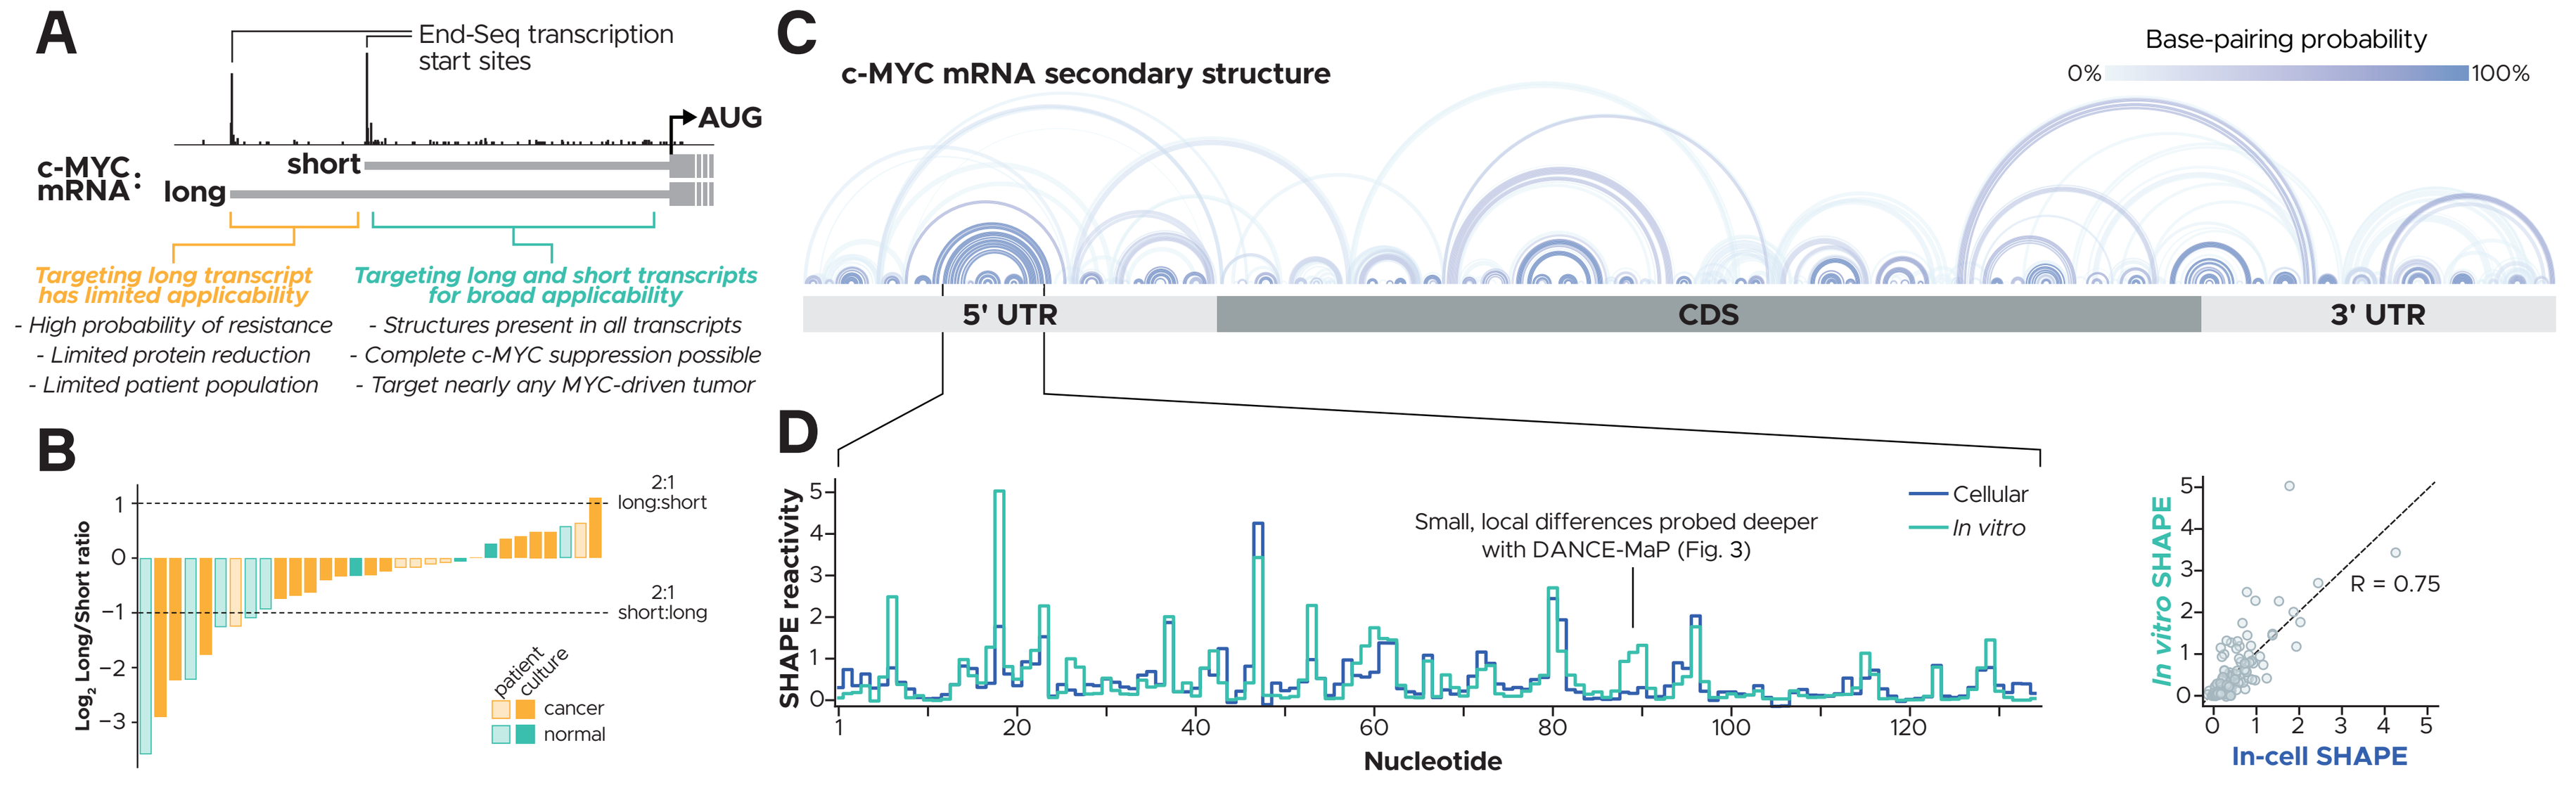
End-Seq transcription start sites
AUG
c-MYC mRNA
short
long
Targeting long transcript has limited applicability
- High probability of resistance
- Limited protein reduction
- Limited patient population
Targeting long and short transcripts for broad applicability
- Structures present in all transcripts
- Complete c-MYC suppression possible
- Target nearly any MYC-driven tumor
Log2 Long/Short ratio
2:1 long:short
2:1 short:long
patient
culture
cancer
normal
c-MYC mRNA secondary structure
Base-pairing probability
0%
100%
5' UTR
CDS
3' UTR
SHAPE reactivity
Small, local differences probed deeper with DANCE-MaP (Fig. 3)
Cellular
In vitro
Nucleotide
R = 0.75
In vitro SHAPE
In-cell SHAPE

Figure 1. Analysis of c-MYC transcription start sites and mRNA secondary structures identifies candidate target structures and highlights strategic importance of targeting both major transcripts. A) c-MYC transcription initiates at one of two transcription start sites (TSS) leading to long and short 5' UTR isoforms. TSS usage is detected and quantified with End-Seq. **B)** End-Seq profiling of patient samples and immortalized cell lines reveal that cancer and normal cells produce a mix of both transcripts. We focused further analyses on the short isoform to ensure all transcripts would contain the target site and to avoid resistance via promoter selection. **C)** RNA secondary structure model of c-MYC mRNA derived from in-cell SHAPE profiling. A well-defined structural element in the 5' UTR was prioritized for further evaluation. **D)** SHAPE reactivity profiles (left) of the 5' UTR element from cellular probing (blue) or an *in vitro* construct (green). Overall correlation between cellular and *in vitro* c-MYC mRNA structures is high (right; Pearson R = 0.75)

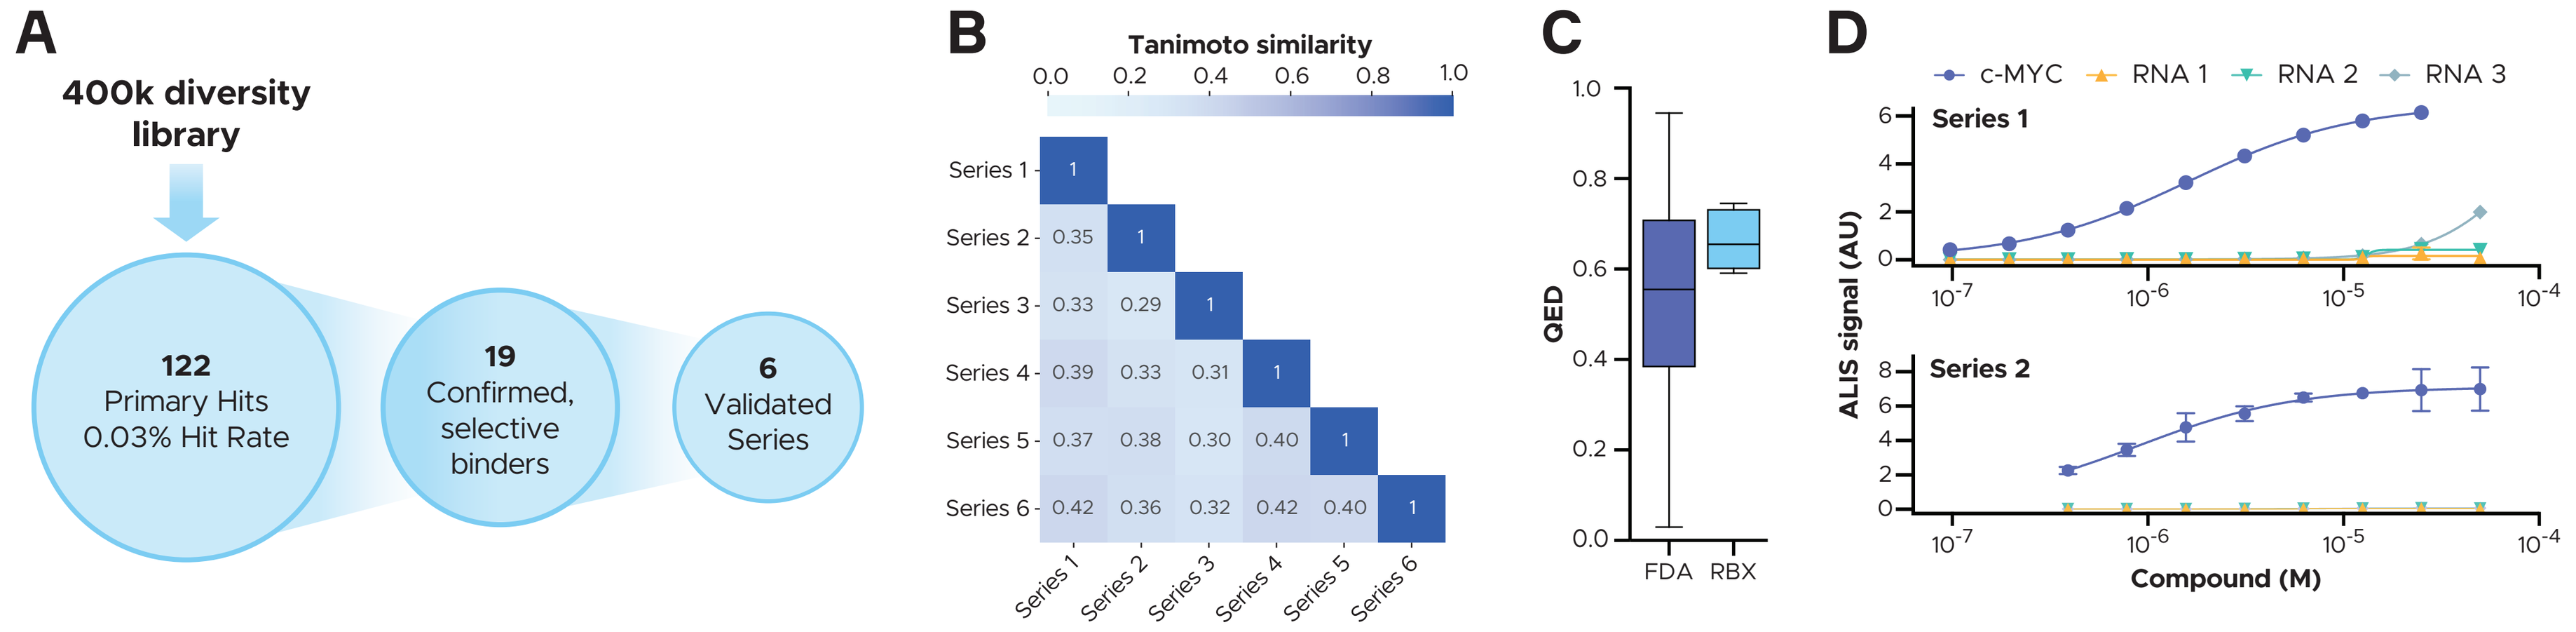
400k diversity library
122
Primary Hits
0.03% Hit Rate
19
Confirmed, selective binders
6
Validated Series
Tanimoto similarity
Series 1
Series 2
Series 3
Series 4
Series 5
Series 6
QED
FDA
RBX
c-MYC
RNA 1
RNA 2
RNA 3
ALIS signal (AU)
Compound (M)

Figure 2. Proprietary Ribometrix high-throughput screen identifies selective c-MYC mRNA-binding compounds. A) High-throughput screen of a 400,000-member diversity library yielded 122 primary hits which were triaged into 6 series with confirmed selective binding. **B)** Pairwise chemical similarity analysis using Tanimoto fingerprints shows that each c-MYC binding series is unique (score < 0.6). **C)** Distributions of QED (quantitative estimate of drug-likeness) scores for 2300 FDA-approved small-molecule drugs and Ribometrix c-MYC series parent molecules. Ribometrix RNA-binding small molecules have chemical properties comparable to approved protein-targeting drugs. **D)** Selectivity analysis of Series 1 (top) and Series 2 (bottom) parent compounds shows strong biophysical selectivity for target c-MYC RNA compared to three highly structured and similarly-sized RNA constructs derived from unrelated human mRNAs.

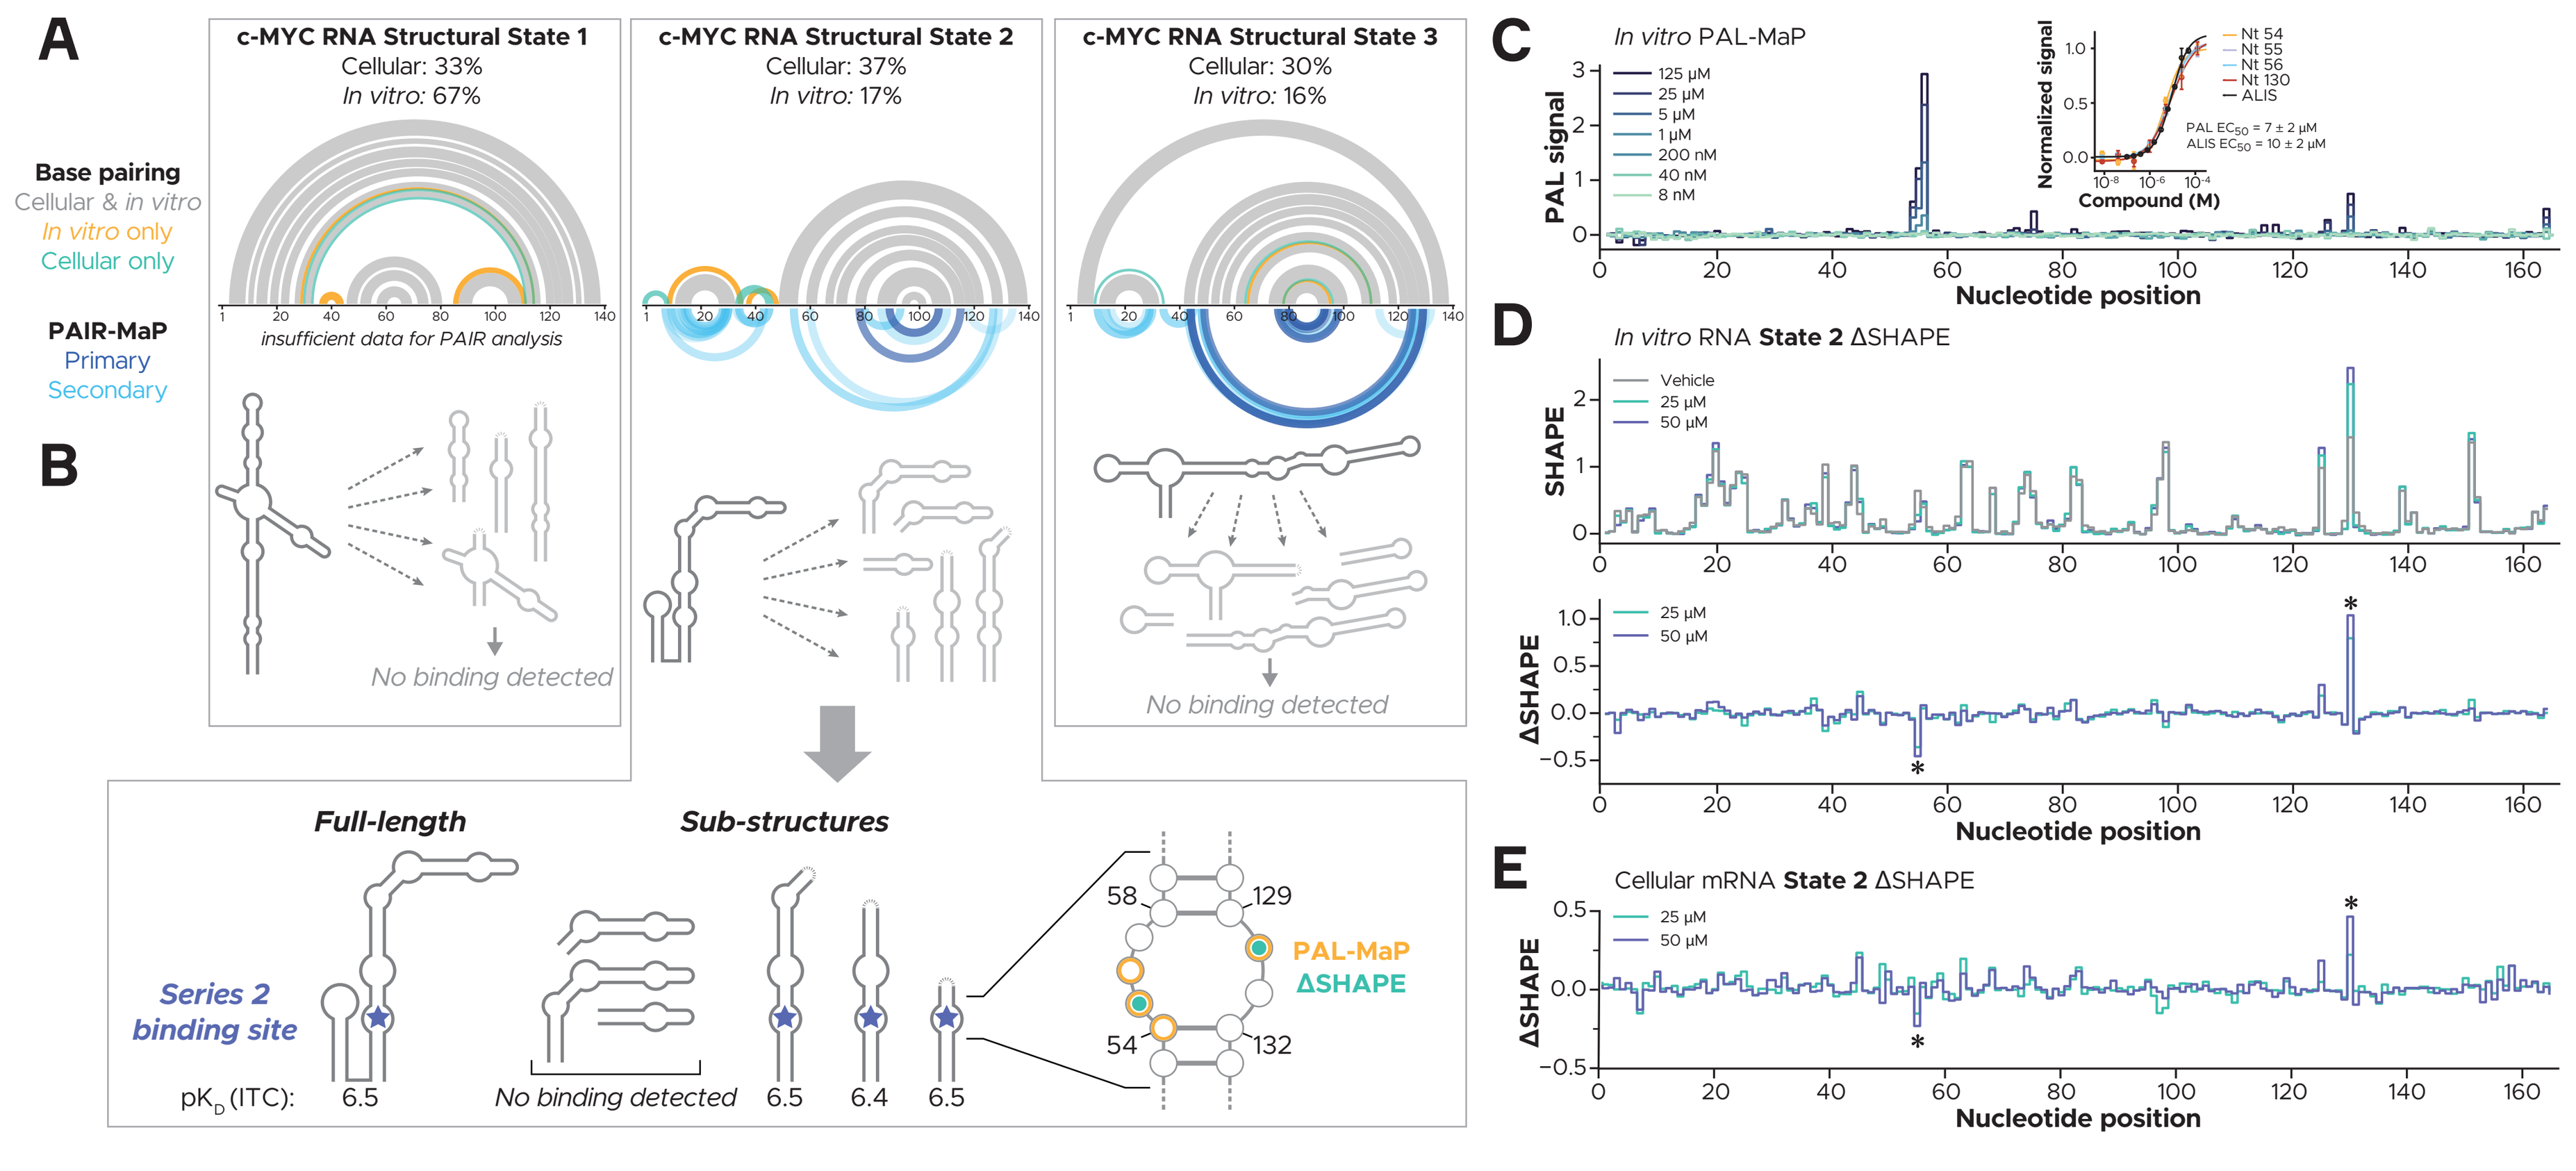
c-MYC RNA Structural State 1
Cellular: 33%
In vitro: 67%
c-MYC RNA Structural State 2
Cellular: 37%
In vitro: 17%
c-MYC RNA Structural State 3
Cellular: 30%
In vitro: 16%
Base pairing
Cellular & in vitro
In vitro only
Cellular only
PAIR-MaP
Primary
Secondary
insufficient data for PAIR analysis
No binding detected
Full-length
Sub-structures
Series 2 binding site
pKD (ITC):
6.5
6.4
PAL-MaP
ΔSHAPE
In vitro PAL-MaP
PAL signal
Normalized signal
Compound (M)
PAL EC50 = 7 ± 2 µM
ALIS EC50 = 10 ± 2 µM
Nucleotide position
In vitro RNA State 2 ΔSHAPE
Vehicle
25 µM
50 µM
SHAPE
ΔSHAPE
Cellular mRNA State 2 ΔSHAPE

Figure 3. Deconvolution of c-MYC RNA structural states and target engagement assays identify Series 2 binding site. A) Cellular and biochemical DANCE- and PAIR-MaP analyses reveal three co-existing RNA conformational states. Top arcs depict minimum free-energy (MFE) conformations of each state. Arc coloring represents base-pairs present in cellular (green), *in vitro* (orange), and both (grey) samples. Bottom arcs depict PAIR-MaP correlations and largely support MFE structures. **B)** MFE structures of each state were used to design sub-constructs for identification of Series 2 compound binding site(s). Only sub-structures arising from State 2 and containing an internal bulge (starred) involving nucleotides 54-58 and 129-132 retained compound binding with potency comparable to full-length RNA. **C)** *In vitro* photoaffinity labeling detected by mutational profiling (PAL-MaP) reveals nucleotides adjacent to the Series 2 binding site. Inset: four nucleotides exhibit a dose response with EC_{50} values comparable to that obtained by ALIS. **D)** SHAPE reactivity profiles for state 2 obtained after *in vitro* compound treatment (top). Significant dose-dependent differences in SHAPE reactivity are observed at positions 55 and 130 (bottom). **E)** In-cell SHAPE probing and DANCE-MaP deconvolution after treatment with Series 2 compound support *in vitro* results and reveals significant changes in SHAPE reactivity at positions 55 and 130. (*$p_{adj} < 0.05$)

RESULTS

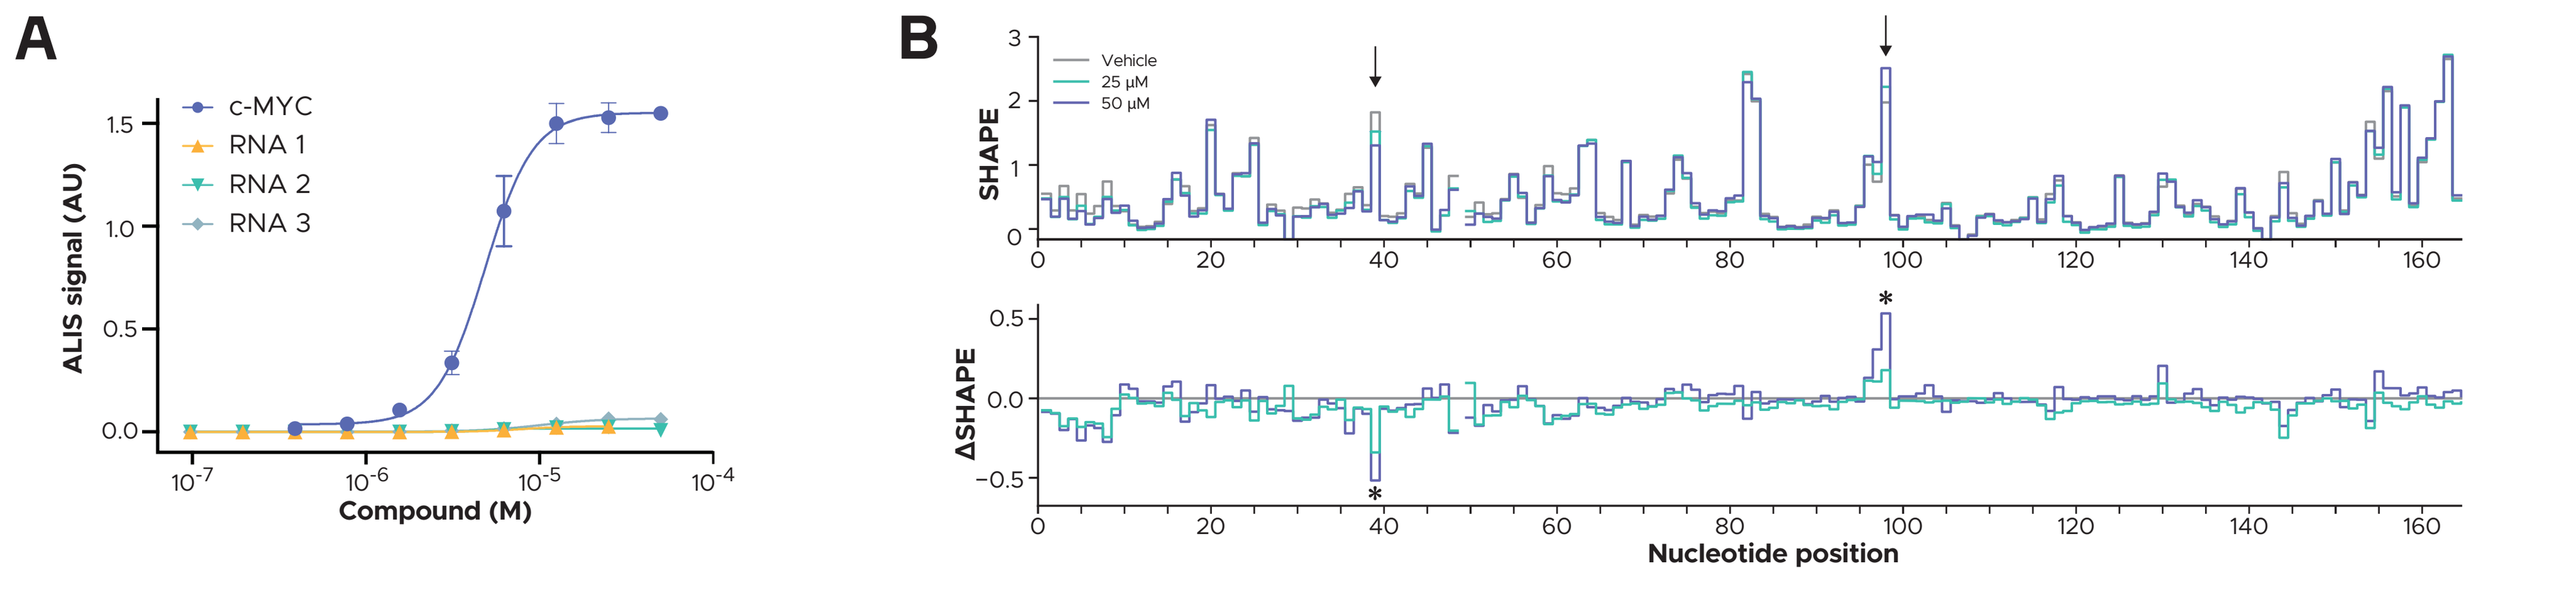
c-MYC
RNA 1
RNA 2
RNA 3
ALIS signal (AU)
Compound (M)
Vehicle
25 µM
50 µM
SHAPE
ΔSHAPE
Nucleotide position

Figure 4. Lead Series 1 engages MYC mRNA in cells. A) *In vitro* selectivity analysis of Series 1 analog RBX-G shows improved selectivity against a panel of well-structured mRNA-derived constructs compared to the parent compound (Fig. 2D). **B)** In-cell ΔSHAPE probing of DMS-273 cells treated for 4 h with RBX-G reveals dose-dependent changes in SHAPE reactivity consistent with direct compound engagement with the cellular mRNA. (*$p_{adj} < 0.05$)

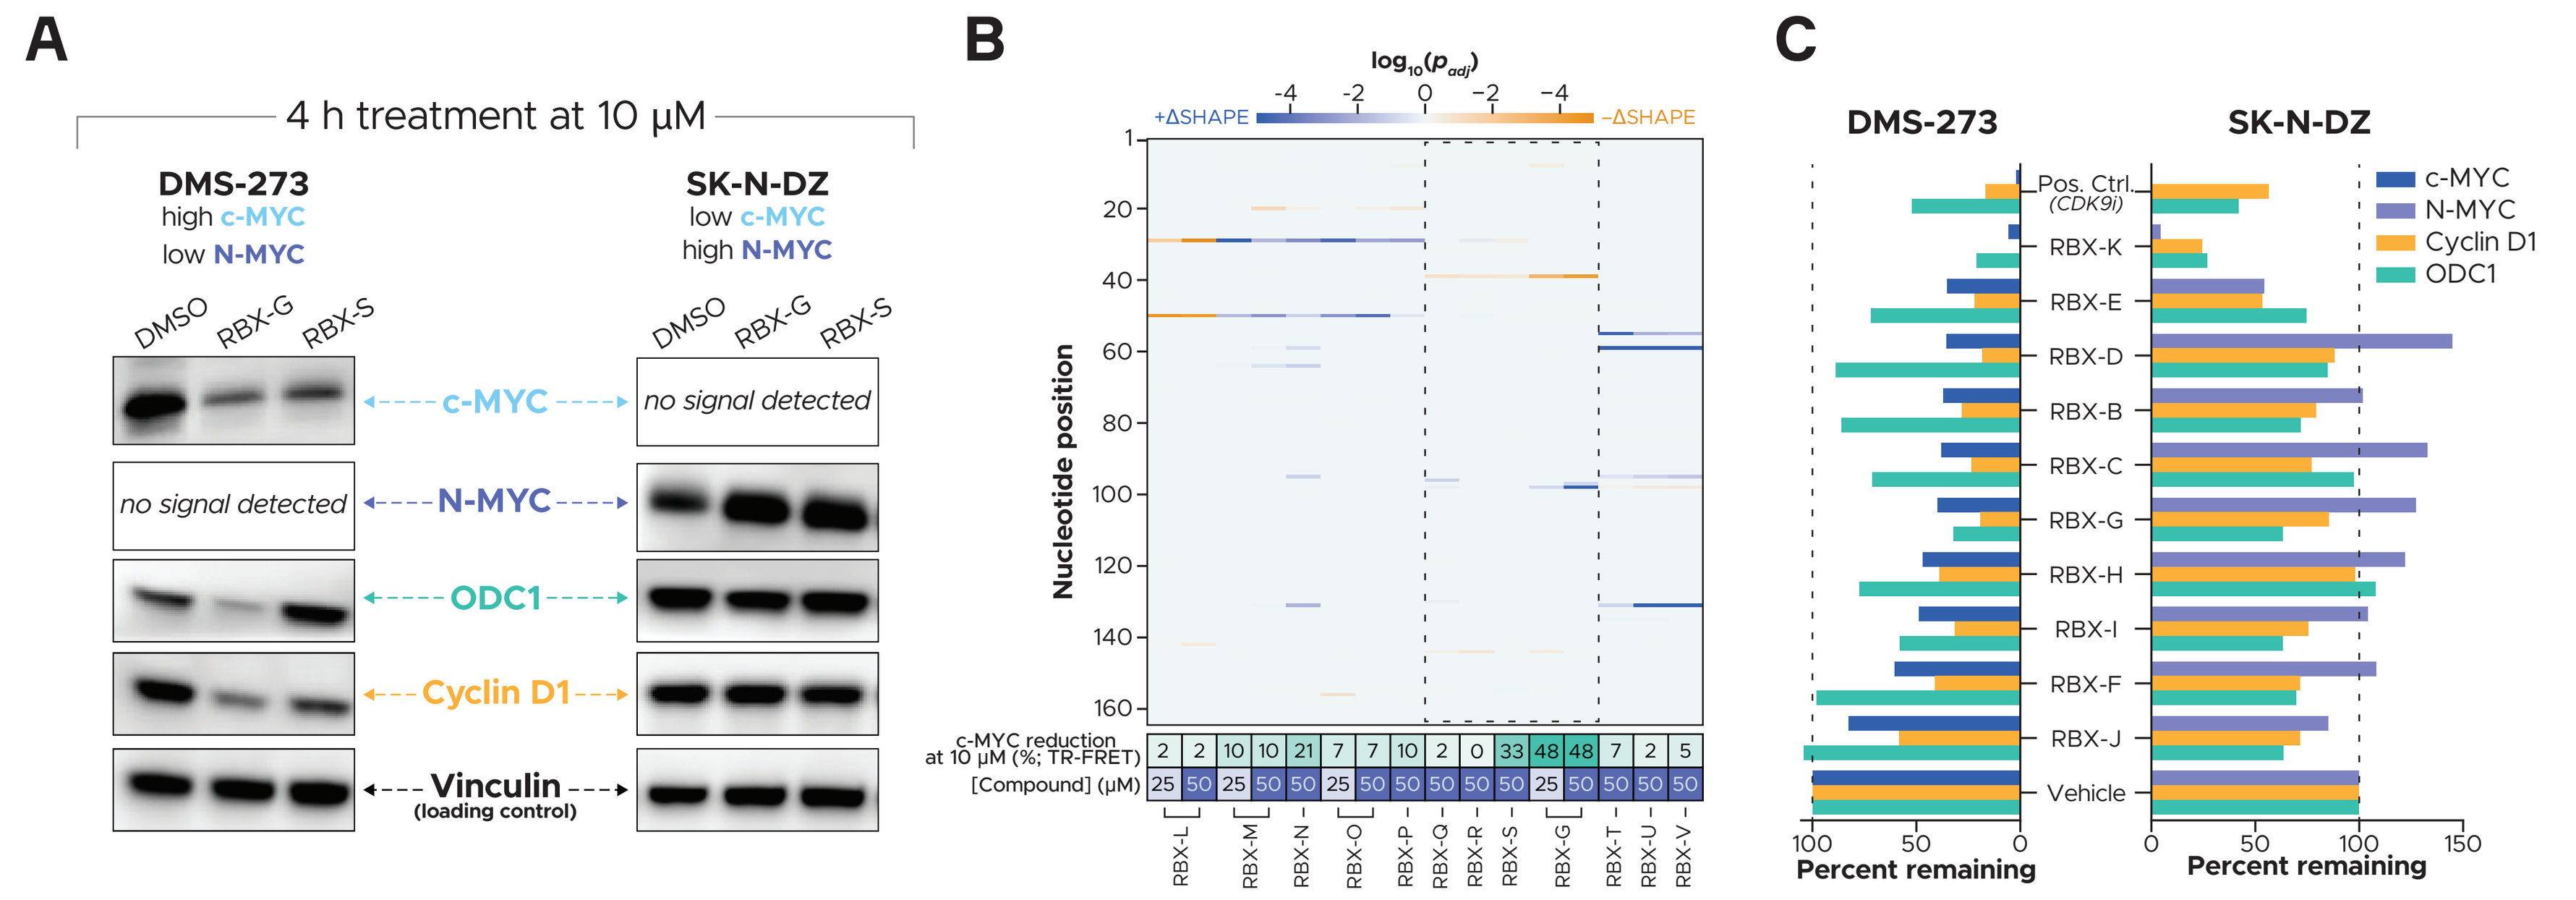
4 h treatment at 10 µM
DMS-273
high c-MYC
low N-MYC
SK-N-DZ
low c-MYC
high N-MYC
DMSO
RBX-G
RBX-S
c-MYC
no signal detected
N-MYC
ODC1
Cyclin D1
Vinculin
(loading control)
log10(padj)
+ΔSHAPE
−ΔSHAPE
Nucleotide position
c-MYC reduction at 10 µM (%; TR-FRET)
[Compound] (µM)
RBX-L
RBX-M
RBX-N
RBX-O
RBX-P
RBX-Q
RBX-R
RBX-S
RBX-G
RBX-T
RBX-U
RBX-V
Pos. Ctrl.
(CDK9i)
RBX-K
RBX-E
RBX-D
RBX-B
RBX-C
RBX-H
RBX-I
RBX-F
RBX-J
Vehicle
c-MYC
N-MYC
Cyclin D1
ODC1
Percent remaining

Figure 5. Lead Series 1 compounds selectively reduce c-MYC and have a distinct ΔSHAPE signature. A) DMS-273 and SK-N-DZ cells (expressing high c-MYC or N-MYC, respectively) were treated with Series 1 compounds RBX-G and RBX-S for 4 h at 10 µM followed by western blot analysis of MYC proteins and two downstream MYC transcriptional targets, ODC1 and Cyclin D1. Reduction of MYC protein and downstream targets was only observed in DMS-273 cells, suggesting that Series 1 compounds act selectively through c-MYC mRNA itself and not through a general MYC pathway mechanism. **B)** In-cell ΔSHAPE probing of DMS-273 cells treated for 4 h with a panel of Series 1 compounds reveals intra-series differences in target engagement. RBX-Q, RBX-R, RBX-S, and RBX-G are among a subset of compounds (dashed box) that exhibit a unique ΔSHAPE pattern involving significant decreases in SHAPE reactivity at position 39. Only compounds within this subset demonstrate selective activity against c-MYC and downstream targets when assayed by western blot. Strong interactions at positions 39 and 98 appear to be critical for stronger reduction of c-MYC protein. **C)** Western blot quantitation from an expanded panel of Series 1 compounds shows that most compounds reduce C-MYC >50% and result in a decrease in Cyclin D1 and ODC1 in DMS-273 cells while N-MYC levels are unaffected or in some cases slightly increased in SK-N-DZ cells.

CONCLUSIONS

- Druggable RNA structure motifs within human mRNAs can be successfully identified and faithfully reproduced *in vitro* for biochemical and biophysical assays.
- RNAs are suitable candidates for biophysical HTS campaigns and yield diverse, selective hits with chemical properties comparable to approved small-molecule drugs.
- Multiple series of RNA-targeting compounds are found to engage the c-MYC cellular mRNA via orthogonal methods and binding sites can be identified even in the context of multiple co-existing structural states.
- Lead Series 1 compounds selectively reduce MYC protein and multiple downstream transcription targets in cells expressing c-MYC but not in cells expressing N-MYC, indicating a c-MYC dependent mechanism rather than general MYC pathway interference.
- Ribometrix c-MYC-targeting small molecules have the potential to address a broad range of oncology indications.
- *In vivo* xenograft studies are anticipated 2H24.

Scan to access online PDF

AACR Annual Meeting, April 5-10, 2024

Contact e-mail: kwarner@ribometrix.com